What will it take to ‘End the HIV epidemic’ in the US? An economic modeling study in 6 cities

Bohdan Nosyk[1,2], Xiao Zang[1,2], Emanuel Krebs[1], Benjamin Enns[1], Jeong E Min[1], Czarina N Behrends[3], Carlos Del Rio[4], Julia C Dombrowski[5], Daniel J Feaster[6], Matthew Golden[5], Brandon DL Marshall[7], Shruti H Mehta[8], Lisa R Metsch[9], Ankur Pandya[10], Bruce R Schackman[3], Steven Shoptaw[11], Steffanie A Strathdee[12] *on behalf of the Localized HIV Economic Modeling Study Group.*

1. BC Centre for Excellence in HIV/AIDS; Vancouver, British Columbia; 2. Faculty of Health Sciences, Simon Fraser University; Vancouver, British Columbia; 3. Department of Healthcare Policy and Research, Weill Cornell Medical College, New York City, New York; 4. Rollins School of Public Health and Emory University School of Medicine, Atlanta, Georgia; 5. Department of Medicine, Division of Allergy and Infectious Disease, University of Washington, Seattle, Washington; 6. Department of Public Health Sciences, Leonard M. Miller School of Medicine, University of Miami, Miami, Florida; 7. School of Public Health, Brown University, Providence, Rhode Island; 8. Department of Epidemiology, Bloomberg School of Public Health, Johns Hopkins University, Baltimore, Maryland; 9. Department of Sociomedical Sciences, Mailman School of Public Health, Columbia University, New York City, New York; 10. Department of Health Policy and Management, Harvard T.H. Chan School of Public Health, Boston, Massachusetts; 11. School of Medicine, University of California Los Angeles, Los Angeles, California; 12. School of Medicine, University of California San Diego, La Jolla, California, .

Poster Number: 01093

Background

- The HIV epidemic in the US is a collection of diverse local microepidemics and targeted strategies have been proposed to reduce HIV incidence by 90% within 10 years.
- We aimed to identify optimal combination implementation strategies of evidence-based interventions to reach these targets in six cities, comprising 24.1% of people living with HIV/AIDS in the US.

Methods

- We used a dynamic, compartmental HIV transmission model to project HIV microepidemics for six US cities: Atlanta, Georgia; Baltimore, Maryland; Los Angeles, California; Miami, Florida; New York City, New York; and Seattle, Washington.
- The adult population aged 15-64 was partitioned by sex at birth, HIV risk group (men who have sex with men [MSM], PWID, MSM who inject drugs [MSMWID] and heterosexuals), race/ethnicity (black/African American, Hispanic/Latinx and non-Hispanic white/others) and sexual risk behavior level (high- vs. low-risk).
- For each city, we calibrated the model to match HIV prevalence, new diagnoses and deaths (2012-2015), and validated against external incidence estimates.
- Interventions were selected from the US Centers for Disease Control and Prevention ‘Compendium of Evidence-Based Interventions and Best Practices for HIV Prevention’ and from the recently published literature (**Table 1**).
- We estimated averted HIV infections and incremental cost-effectiveness ratios (ICERs) (healthcare perspective; 3% annual discount rate, 2018$US), for each intervention and city compared to the status quo over a 20-year time horizon. Interventions were implemented at plausible scale for a 10-year period.

Results

- We assessed 23,040 combinations, with optimal strategies containing between nine (Seattle) and thirteen (Miami) interventions (**Figure 1**).
- Implemented at previously-documented scale-up, these would reduce incidence by 30.7% (95% credible interval: 19.1%-43.7%) (Seattle) to 50.1% (41.5%-58.0%) (NYC) by 2030 (**Figure 2**), at ICERs ranging from cost-saving in Atlanta, Baltimore and Miami to $95,416/QALY in Seattle (**Figure 1**).
- Incidence reductions rose to 39.5% (26.3%-53.8%) in Seattle to 83.6% (70.8%-87.0%) in Baltimore at ideal implementation (**Figure 2**).

- Funding by the **National Institutes on Drug Abuse**, (R01-DA041747).

Within these six cities, implementing combinations of evidence-based interventions can ***provide good value*** and ***approach national incidence reduction targets*** in some settings.

Table 1. Selected evidence-based interventions, their effectiveness and previously-documented scale-up of delivery based on the RE-AIM framework

	Effectiveness and target population¶ [Evidence Level*]	Population Coverage† Previously-documented Implementation	Population Coverage† Ideal Implementation**
Protect: HIV prevention programs			
Syringe service program (SSP)	Clean injection equipment reduces parenteral HIV transmission by 58% among PWID. [2a]	Additional scale-up of 11%	90%
MOUD with buprenorphine	Office-based MOUD for PWID reduces the number of injections by 54%. [2a]	Additional scale-up of 14%	90%
MOUD with methadone	OTP-based MOUD for PWID reduces the number of injections by 54%. [2a]	Additional scale-up of 12%	90%
Targeted PrEP for high-risk MSM	PrEP reduces the risk of HIV infection by 60%Ŧ among high-risk MSM and MWID.§ [1b]	Additional scale-up of 68%	90%
Diagnose: HIV Testing			
Opt-out testing in ER	Routine HIV testing increases by 28% among individuals visiting the ER. [1b]	3%-6%	10%-26%
Opt-out testing in primary care	Routine HIV testing increases by 28% during primary care visits. [1b]	25%-40%	53%-85%
EMR testing offer reminder	HIV testing increases by 178% among individuals visiting the ER. [2b]	11%-29%	10%-26%
Nurse-initiated rapid testing	HIV testing increases by 73% during health care visits.^ [2b]	25%-40%	53%-85%
MOUD integrated rapid testing	On-site HIV testing increases by 352% among PWID receiving MOUD. [1b]	17%	49%
Treat: ART engagement and re-engagement			
Case management (ARTAS)	ART initiation increases by 41% among PLHIV linked to care. [1b]	57%	77%
Care coordination	ART retention increases by 10% among PLHIV. [2b]	10%-20%	34%-68%
Targeted care coordination	ART retention increases by 32% among PLHIV with CD4<200 cells per μL. [2b]	30%-46%	41%-63%
EMR ART engagement reminder	ART drop-out is reduced by 31% among PLHIV on ART. [1b]	42%-78%	54%-82%
RAPID ART initiation	Immediate ART initiation increases by 32% among newly diagnosed PLHIV. [3b]	30%-61%	41%-84%
Enhanced personal contact	ART re-initiation increases by 22% among PLHIV having dropped-out of ART. [1b]	45%	62%
Re-linkage program	ART re-initiation increases by 70% among PLHIV who are out-of-care. [2b]	8%	22%

PWID: People who inject drugs; MOUD: Medication for opioid use disorder; OTP: Opioid treatment program; PrEP: Pre-exposure prophylaxis; MSM: Men who have sex with men; MWID: MSM PWID; ER: Hospital emergency room; EMR: Electronic medical records; PLHIV: People living with HIV; ART: antiretroviral therapy.

¶ Interventions target the general adult population 15-64 unless noted otherwise.

† Where applicable, scale ranges indicate evidence stratified by sex/gender and/or race/ethnicity and/or city/region.

* Adapted from the Oxford Centre for Evidence-based Medicine – Levels of Evidence: 1a - Systematic review of RCTs; 1b - Individual high-quality RCT; 2a - Systematic review of cohort studies; 2b - Individual cohort study or quasi-experimental study; 3a - Systematic review of case-control studies; 3b - Individual case-control study; 4 - Case series; 5- Expert opinion.

** Ideal implementation defined as 90% adoption.

Ŧ Effectiveness: efficacy for 4 doses/week [96% (90%, 99%)][19] X protective level adherence [62.5% (≥4 doses/week)][20].

§ We assumed that 25% of MSM are high-risk and indicated for PrEP in accordance with CDC guidelines[21].

^ With either a physician or health care professional.

Conclusion

- Within these six cities, implementing combinations of evidence-based interventions can provide good value and approach national incidence reduction targets in some settings, however, complementary strategies to overcome social and structural barriers to HIV care will be required to reach national ‘Ending the HIV epidemic’ targets by 2030.

Figure 1. City-level health production functions comprising optimal combinations of interventions for different investment levels

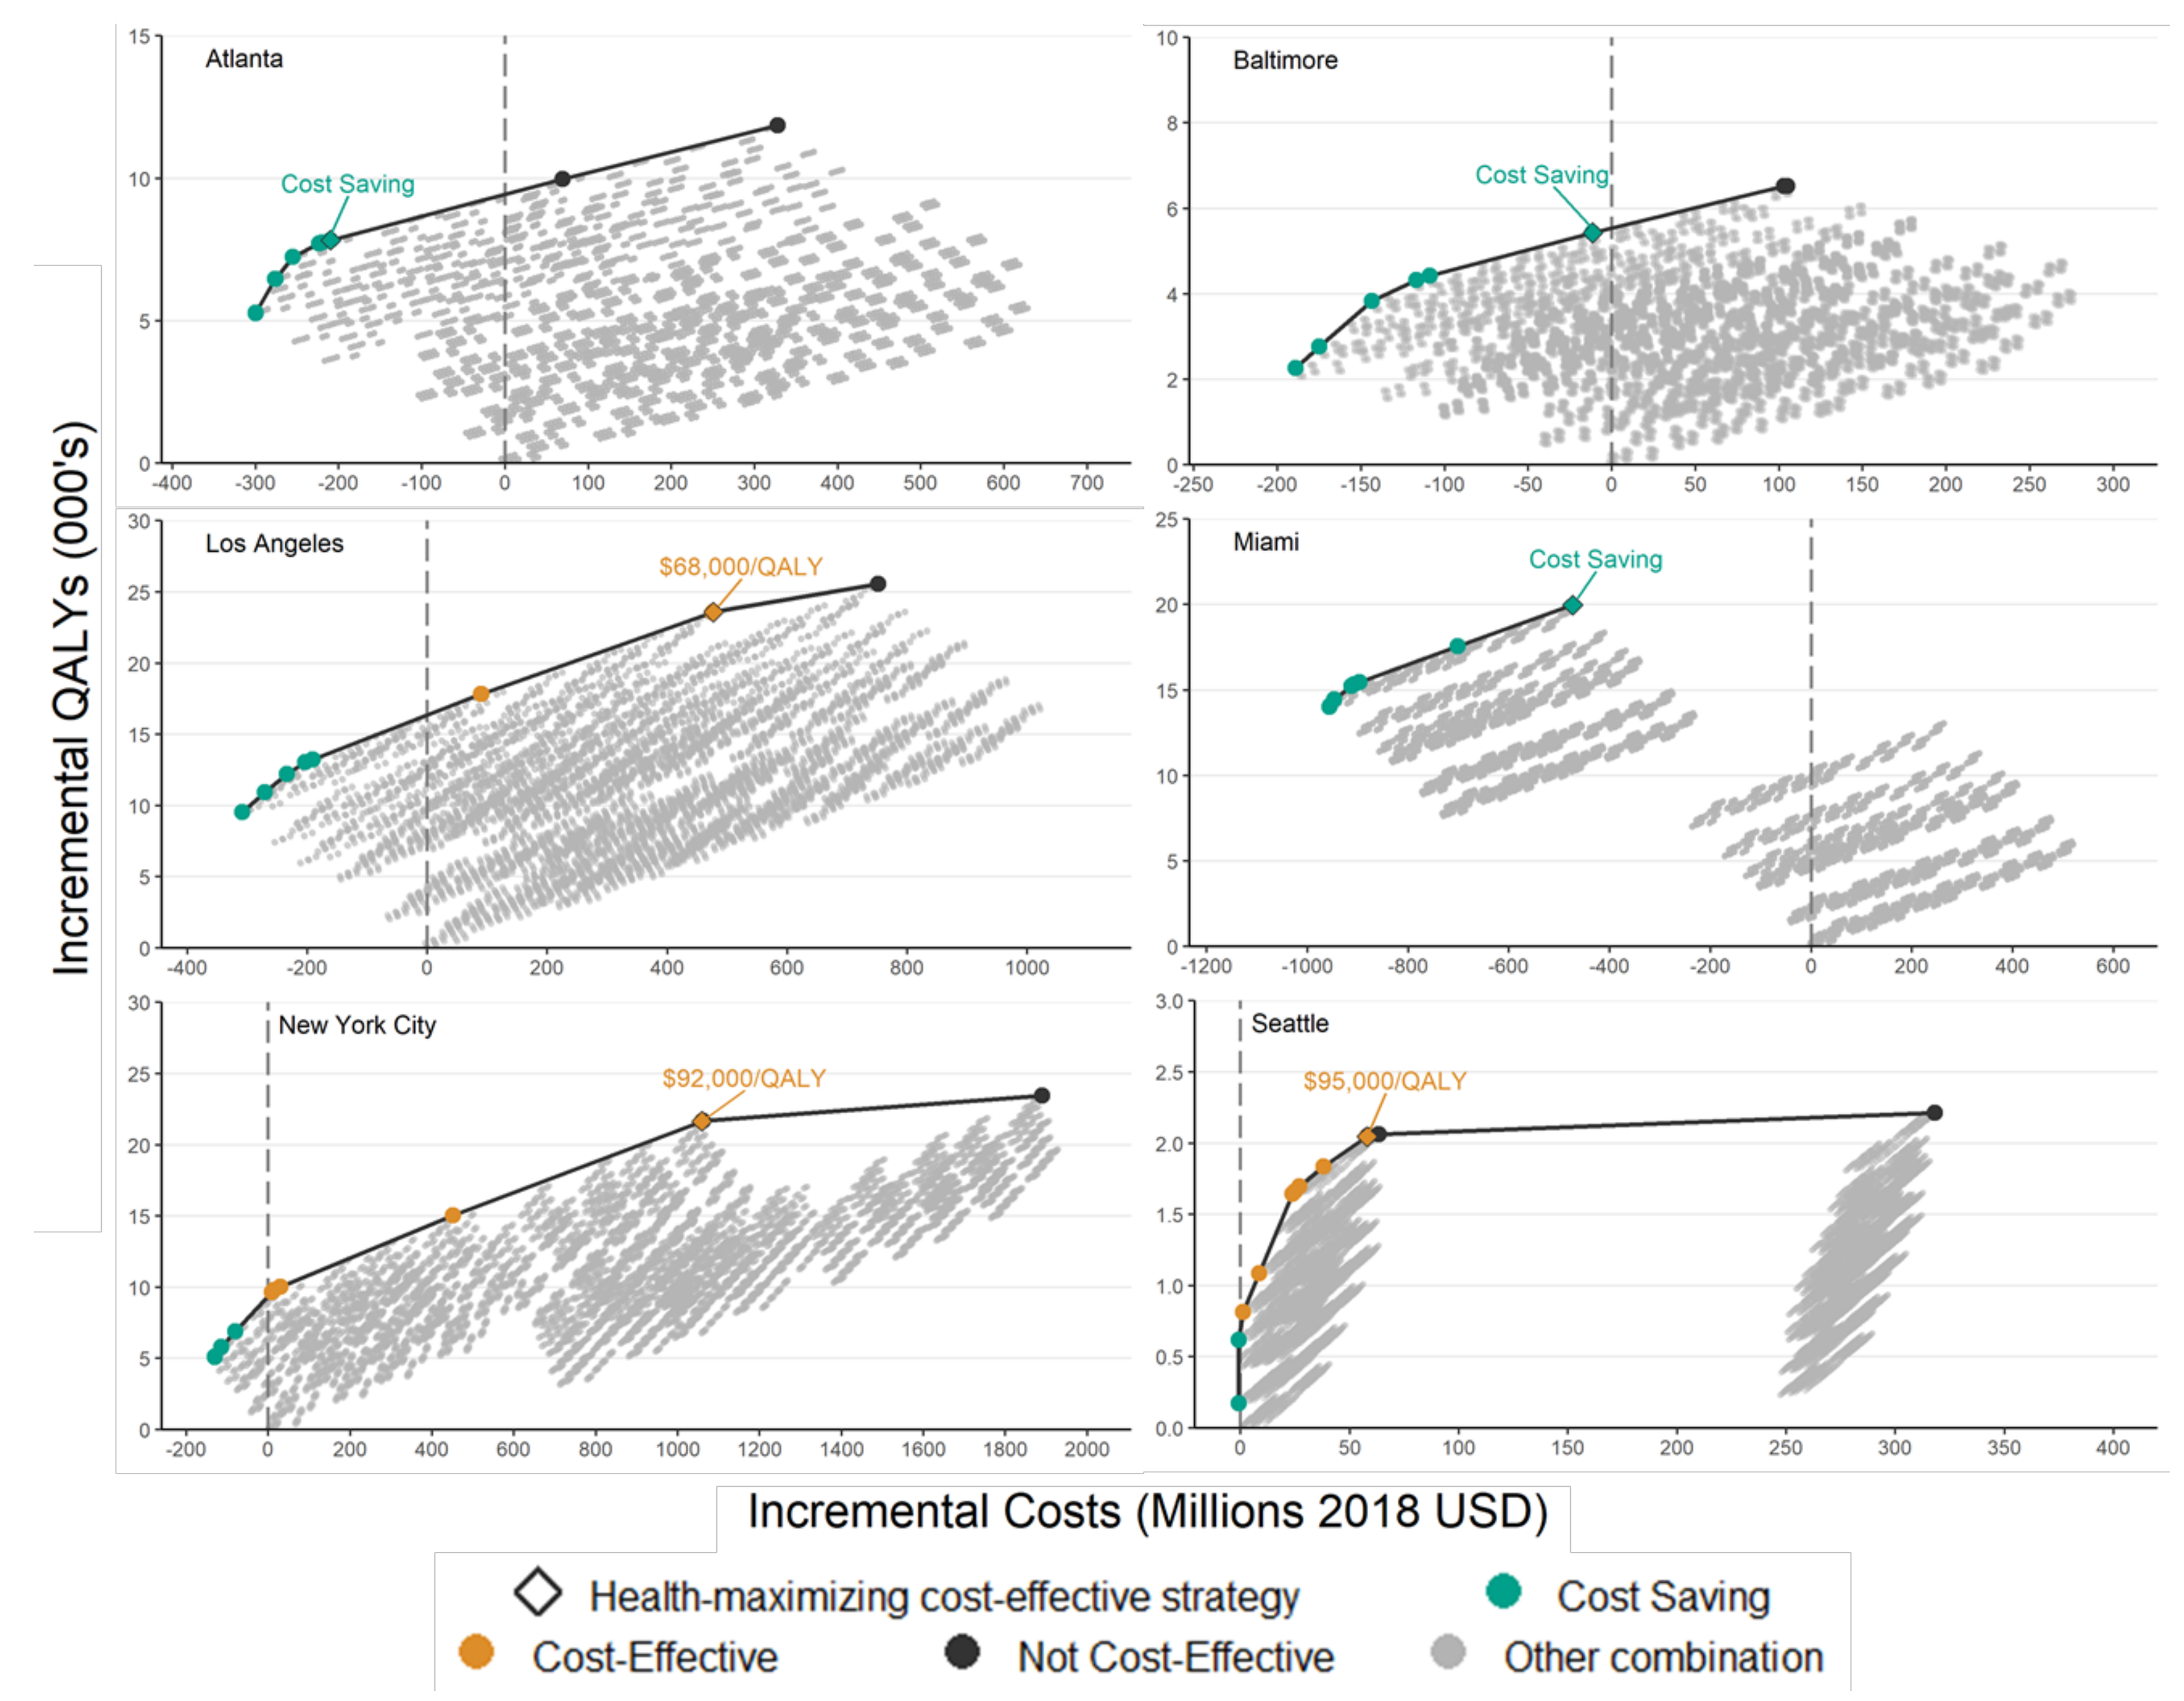

Atlanta
Baltimore
Los Angeles
Miami
New York City
Seattle
Cost Saving
$68,000/QALY
$92,000/QALY
$95,000/QALY
Incremental QALYs (000's)
Incremental Costs (Millions 2018 USD)
Health-maximizing cost-effective strategy
Cost Saving
Cost-Effective
Not Cost-Effective
Other combination

Figure 2. Projected reductions in HIV incidence at status quo service levels compared to implementation of optimal strategies at previously documented scale-up and ideal implementation

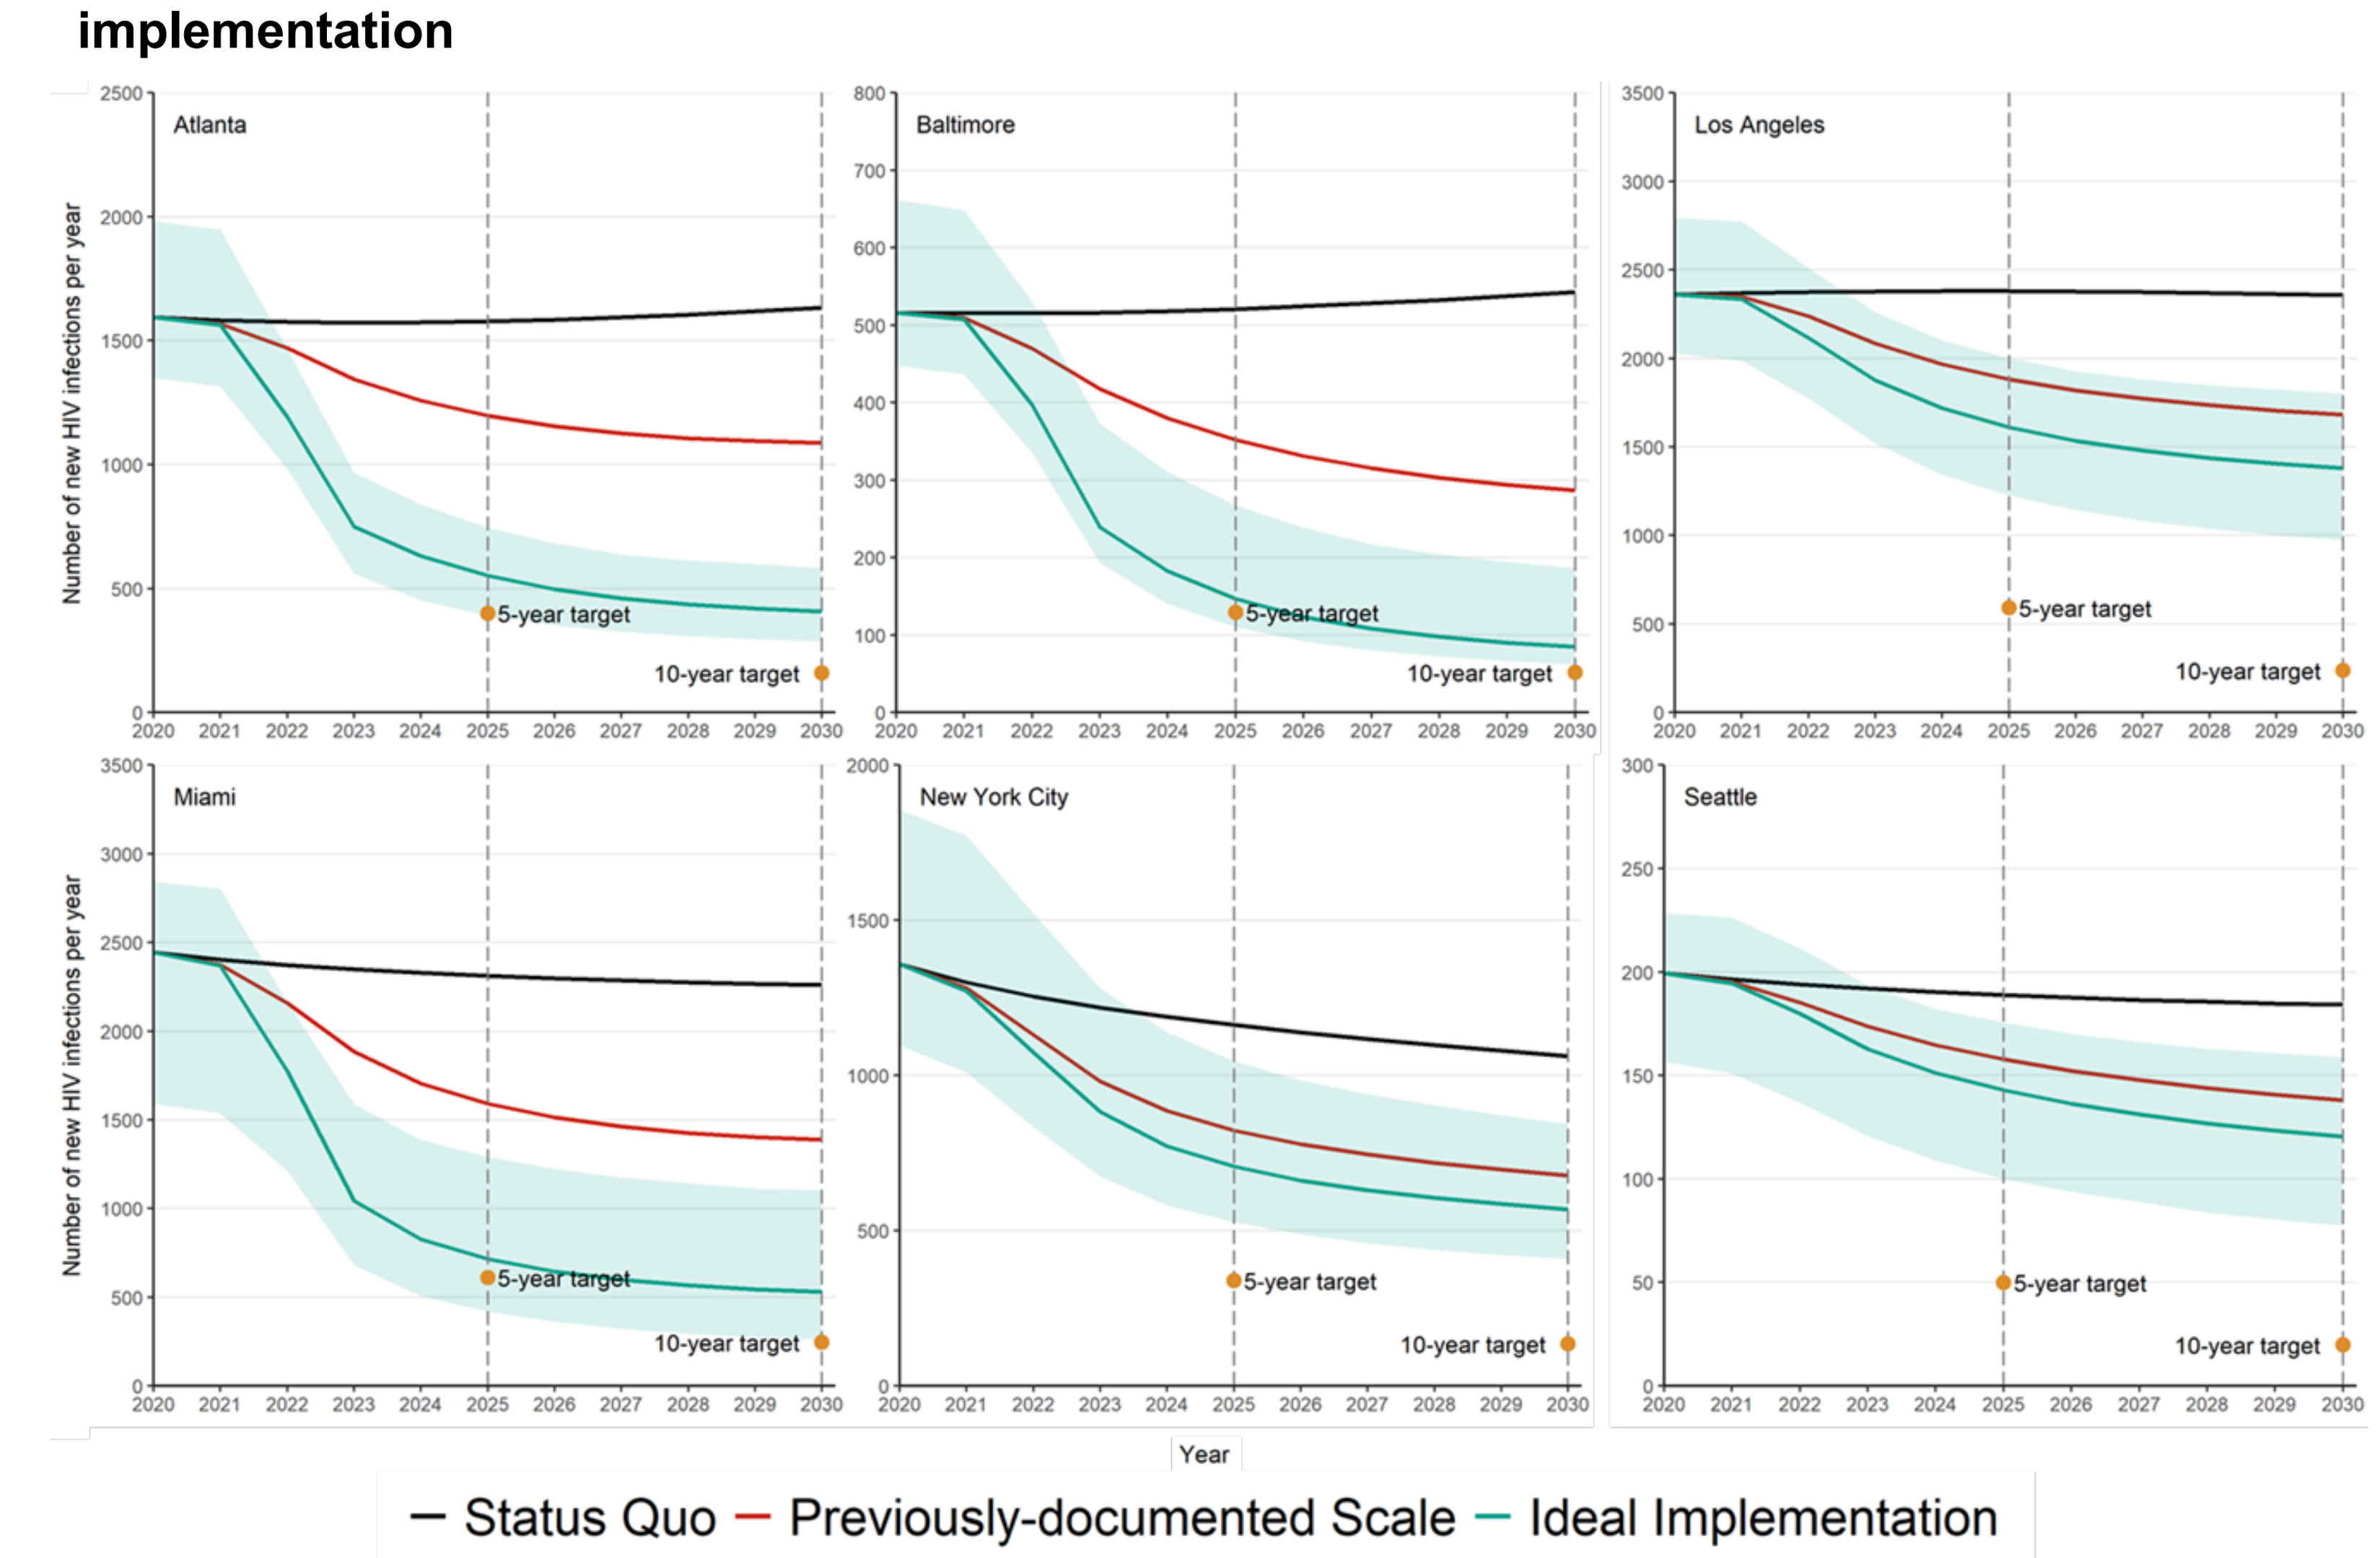

Atlanta
Baltimore
Los Angeles
Miami
New York City
Seattle
Number of new HIV infections per year
5-year target
10-year target
Year
Status Quo
Previously-documented Scale
Ideal Implementation