Ending the HIV epidemic among persons who inject drugs: a cost-effectiveness analysis in six U.S. cities

Emanuel Krebs[1], Xiao Zang[1,2], Benjamin Enns[1], Jeong E Min[1], Czarina N Behrends[3], Carlos Del Rio[4], Julia C Dombrowski[5], Daniel J Feaster[6], Kelly A Gebo[7], Brandon DL Marshall[8], Shruti H Mehta[9], Lisa R Metsch[10], Ankur Pandya[11], Bruce R Schackman[3], Steffanie A Strathdee[12], Bohdan Nosyk[1,2] *on behalf of the Localized HIV Economic Modeling Study Group.*

1. BC Centre for Excellence in HIV/AIDS; Vancouver, British Columbia; 2. Faculty of Health Sciences, Simon Fraser University; Vancouver, British Columbia; 3. Department of Healthcare Policy and Research, Weill Cornell Medical College, New York City, New York; 4. Rollins School of Public Health and Emory University School of Medicine, Atlanta, Georgia; 5. Department of Medicine, Division of Allergy and Infectious Disease, University of Washington, Seattle, Washington; 6. Department of Public Health Sciences, Leonard M. Miller School of Medicine, University of Miami, Miami, Florida; 7. School of Medicine, Bloomberg School of Public Health, Johns Hopkins University, Baltimore, Maryland; 8. School of Public Health, Brown University, Providence, Rhode Island; 9. Department of Epidemiology, Bloomberg School of Public Health, Johns Hopkins University, Baltimore, Maryland; 10. Department of Sociomedical Sciences, Mailman School of Public Health, Columbia University, New York City, New York; 11. Department of Health Policy and Management, Harvard T.H. Chan School of Public Health, Boston, Massachusetts; 12. School of Medicine, University of California San Diego, La Jolla, California.

Poster Number: 01144

Background

- In the United States, persons who inject drugs (PWID) continue to be disproportionately at risk of HIV infection.
- There is considerable evidence suggesting that implementation of prevention programs can be highly effective in reducing transmission of HIV among PWID, and that HIV incidence among PWID can be reduced substantially with focused, locally-oriented strategies in treating and preventing HIV.
- We aimed to determine the highest-valued combination implementation strategies to reduce the burden of HIV among PWID in six US cities.

Methods

- We used a previously validated dynamic, compartmental HIV transmission model to project distinct HIV microepidemics for six US cities: Atlanta, Georgia; Baltimore, Maryland; Los Angeles, California; Miami, Florida; New York City, New York; and Seattle, Washington.
- In each city, the adult population aged 15-64 was partitioned by sex at birth, HIV risk group (men who have sex with men [MSM], PWID, MSM who inject drugs [MSMWID] and heterosexuals), race/ethnicity (black/African American, Hispanic/Latinx and non-Hispanic white/others) and sexual risk behavior level (high- vs. low-risk).
- For each city, we calibrated the model to match HIV prevalence, new diagnoses and deaths (2012-2015), and validated against external incidence estimates.
- We estimated averted HIV infections and incremental cost-effectiveness ratios (ICERs) (healthcare perspective; 3% annual discount rate, 2018$US), for each intervention and city compared to the status quo over a 20-year time horizon. Interventions were implemented at plausible scale for a 10-year period.

Results

- Selected strategies included between six (Atlanta and Seattle) and twelve (Miami) individual interventions (**Figure 1**). Care coordination to improve ART engagement and RAPID ART were not included in any city's optimal strategy while expanded access to MOUD and rapid HIV testing integrated with MOUD were included in all.
- HIV incidence reductions of 8.1% [2.8%–13.2%] (Seattle) to 54.4% [37.6%–73.9%] (Miami) by 2030 (Figure 3), and with large reductions in Miami, Los Angeles and Atlanta (75.5%, 49.0% and 44.8% respectively) and Baltimore, New York City and Seattle reaching 16.1%, 17.7% and 19.2% reductions, respectively, when implemented at near-ideal levels (**Figure 2**).

Evidence-based ***interventions targeted to PWID*** can deliver ***considerable value***; however, ending the HIV epidemic among PWID will require innovative implementation and support to ***reduce social and structural barriers to care***.

Table 1. Description, effectiveness and scale-up implementation scenarios for the evidence-based HIV prevention programs and care interventions included in our analysis.

Intervention	Supporting evidence			Description and effectiveness**	Scale-up implementation scenarios†	
	Source [Evidence Level*]	Study Design	Study Setting		Optimistic	Ideal^
HIV prevention programs						
Syringe service programs (SSP)	Aspinall et al. 2014 Int J Epi [2a]	Meta-analysis	SSP	Clean injection equipment reduces the risk of parenteral HIV transmission by 58%.	200 syringes / PWID / year ‡	90%
MOUD with buprenorphine	MacArthur et al. 2012 BMJ [2a]	Meta-analysis	Primary Care & OTP	Office-based MOUD reduces the number of shared injections by 54% for PWID with OUD.§	29% #	90% ##
MOUD with methadone	MacArthur et al. 2012 BMJ [2a]	Meta-analysis	Primary Care & OTP	Opioid treatment program-based MOUD reduces the number of shared injections by 54% for PWID with OUD.§	Additional scale-up of 23%	90% ##
Full-time PrEP	Liu et al. 2016 JAMA Intern Med	RCT substudy & Cohort study	Primary Care	Protective level adherence to PrEP (≥4 doses/week) reduces the risk of HIV infection by 60%.₸	50%	90%
HIV Testing						
EMR testing offer reminder	Felsen et al. 2017 JAIDS [2b]	Quasi-exp. pre/post	Hospital	HIV testing increases by 178% among among PWID visiting the ER.	13%-35%	14%-36% ^^
Nurse-initiated rapid testing	Anaya et al. 2008 J Gen Intern Med [2b]	RCT	Primary Care	Nurse-initiated screening and rapid testing increases HIV testing by 73% during health care visits.	34%-52%	56%-87%
MOUD integrated rapid testing	Metsch et al. 2012 Am J Pub H [1b]	RCT	DTP	On-site rapid testing increases HIV testing by 352% among PWID receiving MOUD.	22%	49%
ART engagement						
Case management (ARTAS)	Gardner et al. 2005 AIDS [1b]	RCT	HIV clinics	Contacts with a case manager increases ART initiation by 41% among PLHIV linked to care.	61%	77%
Care coordination	Robertson et al. 2018 Am J Epi [2b]	Pre/post ǁ	HIV clinics	Comprehensive care coordination increases ART retention by 10% among PLHIV.	12%-25%	34%-68%
Targeted care coordination	Robertson et al. 2018 Am J Epi [2b]	Pre/post ǁ	HIV clinics	Targeted comprehensive care coordination increases ART retention by 32% among PLHIV with CD4<200 cells per μL.	41%-48%	57%-66%
EMR ART engagement reminder	Robbins et al. 2012 Ann Int Med [1b]	RCT	HIV clinics	Interactive EMR alerts reduces ART drop-out by 31% among PLHIV on ART.	47%-84%	60%-91% ^^
RAPID ART initiation	Pilcher et al. 2017 JAIDS [3b]	Cohort study	HIV clinics	Multidisciplinary care and support increases immediate ART initiation by 32% among newly diagnosed PLHIV.	38%-71%	47%-90%
ART re-engagement						
Enhanced personal contact	Gardner et al. 2014 Clin Infect Dis [1b]	RCT	HIV clinics	Continuous contact increases ART re-initiation by 22% among PLHIV having dropped-out of ART.	49%	62%
Re-linkage program	Bove et al. 2015 JAIDS [2b]	Cohort study	HIV clinics	Outreach using surveillance data increases ART re-initiation by 70% among PLHIV who are out-of-care.	10%	22%

PWID: Pepople who inject drugs; OUD: opioid use disorder; MOUD: Medication for OUD; PrEP: Pre-exposure prophylaxis; EMR: Electronic medical records; ER: Hospital emergency room; PLHIV: People living with HIV; RAPID: Rapid ART Program for Individuals with an HIV Diagnosis; RCT: Randomized control trial.
* Levels of evidence adapted from Oxford Centre for Evidence-based Medicine – Levels of Evidence: 1a - Systematic review of RCTs; 1b - Individual high-quality RCT; 2a - Systematic review of cohort studies; 2b - Individual cohort study or quasi-experimental study; 3a - Systematic review of case-control studies; 3b - Individual case-control study; 4 - Case series.
** Interventions target the PWID adult population 15-64 including men who have sex with men who inject drugs.
^ Ideal implementation refers to 90% adoption unless otherwise noted by ^^ which refers to 100% adoption of EMR.
§ MOUD also reduces the risk of mortality, increases quality of life, and decreases the probability of ART discontinuation.
† Where applicable, scale-up ranges indicate evidence stratified by sex/gender and/or race/ethnicity and/or city/region.
‡ As recommended by the World Health Organization (WHO) [29], except Seattle (400 syringes / PWID / year) since status quo service levels were already equivalent to this level.
As recommended by the World Health Organization (WHO) [29], 40% coverage among the 72.7% of PWID with an OUD [26] results in 29% coverage among all PWID.
Maximum 90% coverage of both medications combined among the 72.7% of PWID with an OUD [26].
₸ Effectiveness defined as efficacy for 4 doses/week [96% (90%, 99%)] X protective level adherence [62.5% (associated with taking ≥4 doses/week)], further details in the supplement.
ǁ Study with contemporaneous surveillance registry–based comparison group

Conclusion

- Distinct combinations of evidence-based interventions targeted to PWID were required to produce the greatest public health impact in each setting.
- Evidence-based interventions targeted to PWID can deliver considerable value, however ending the HIV epidemic among PWID will require innovative implementation strategies and supporting programs to reduce social and structural barriers to care.
- Funding by the **National Institutes on Drug Abuse**, (R01-DA041747).

Figure 1. City-level health production functions for evidence-based prevention and care interventions targeted to persons who inject drugs and men who have sex with men who inject drugs

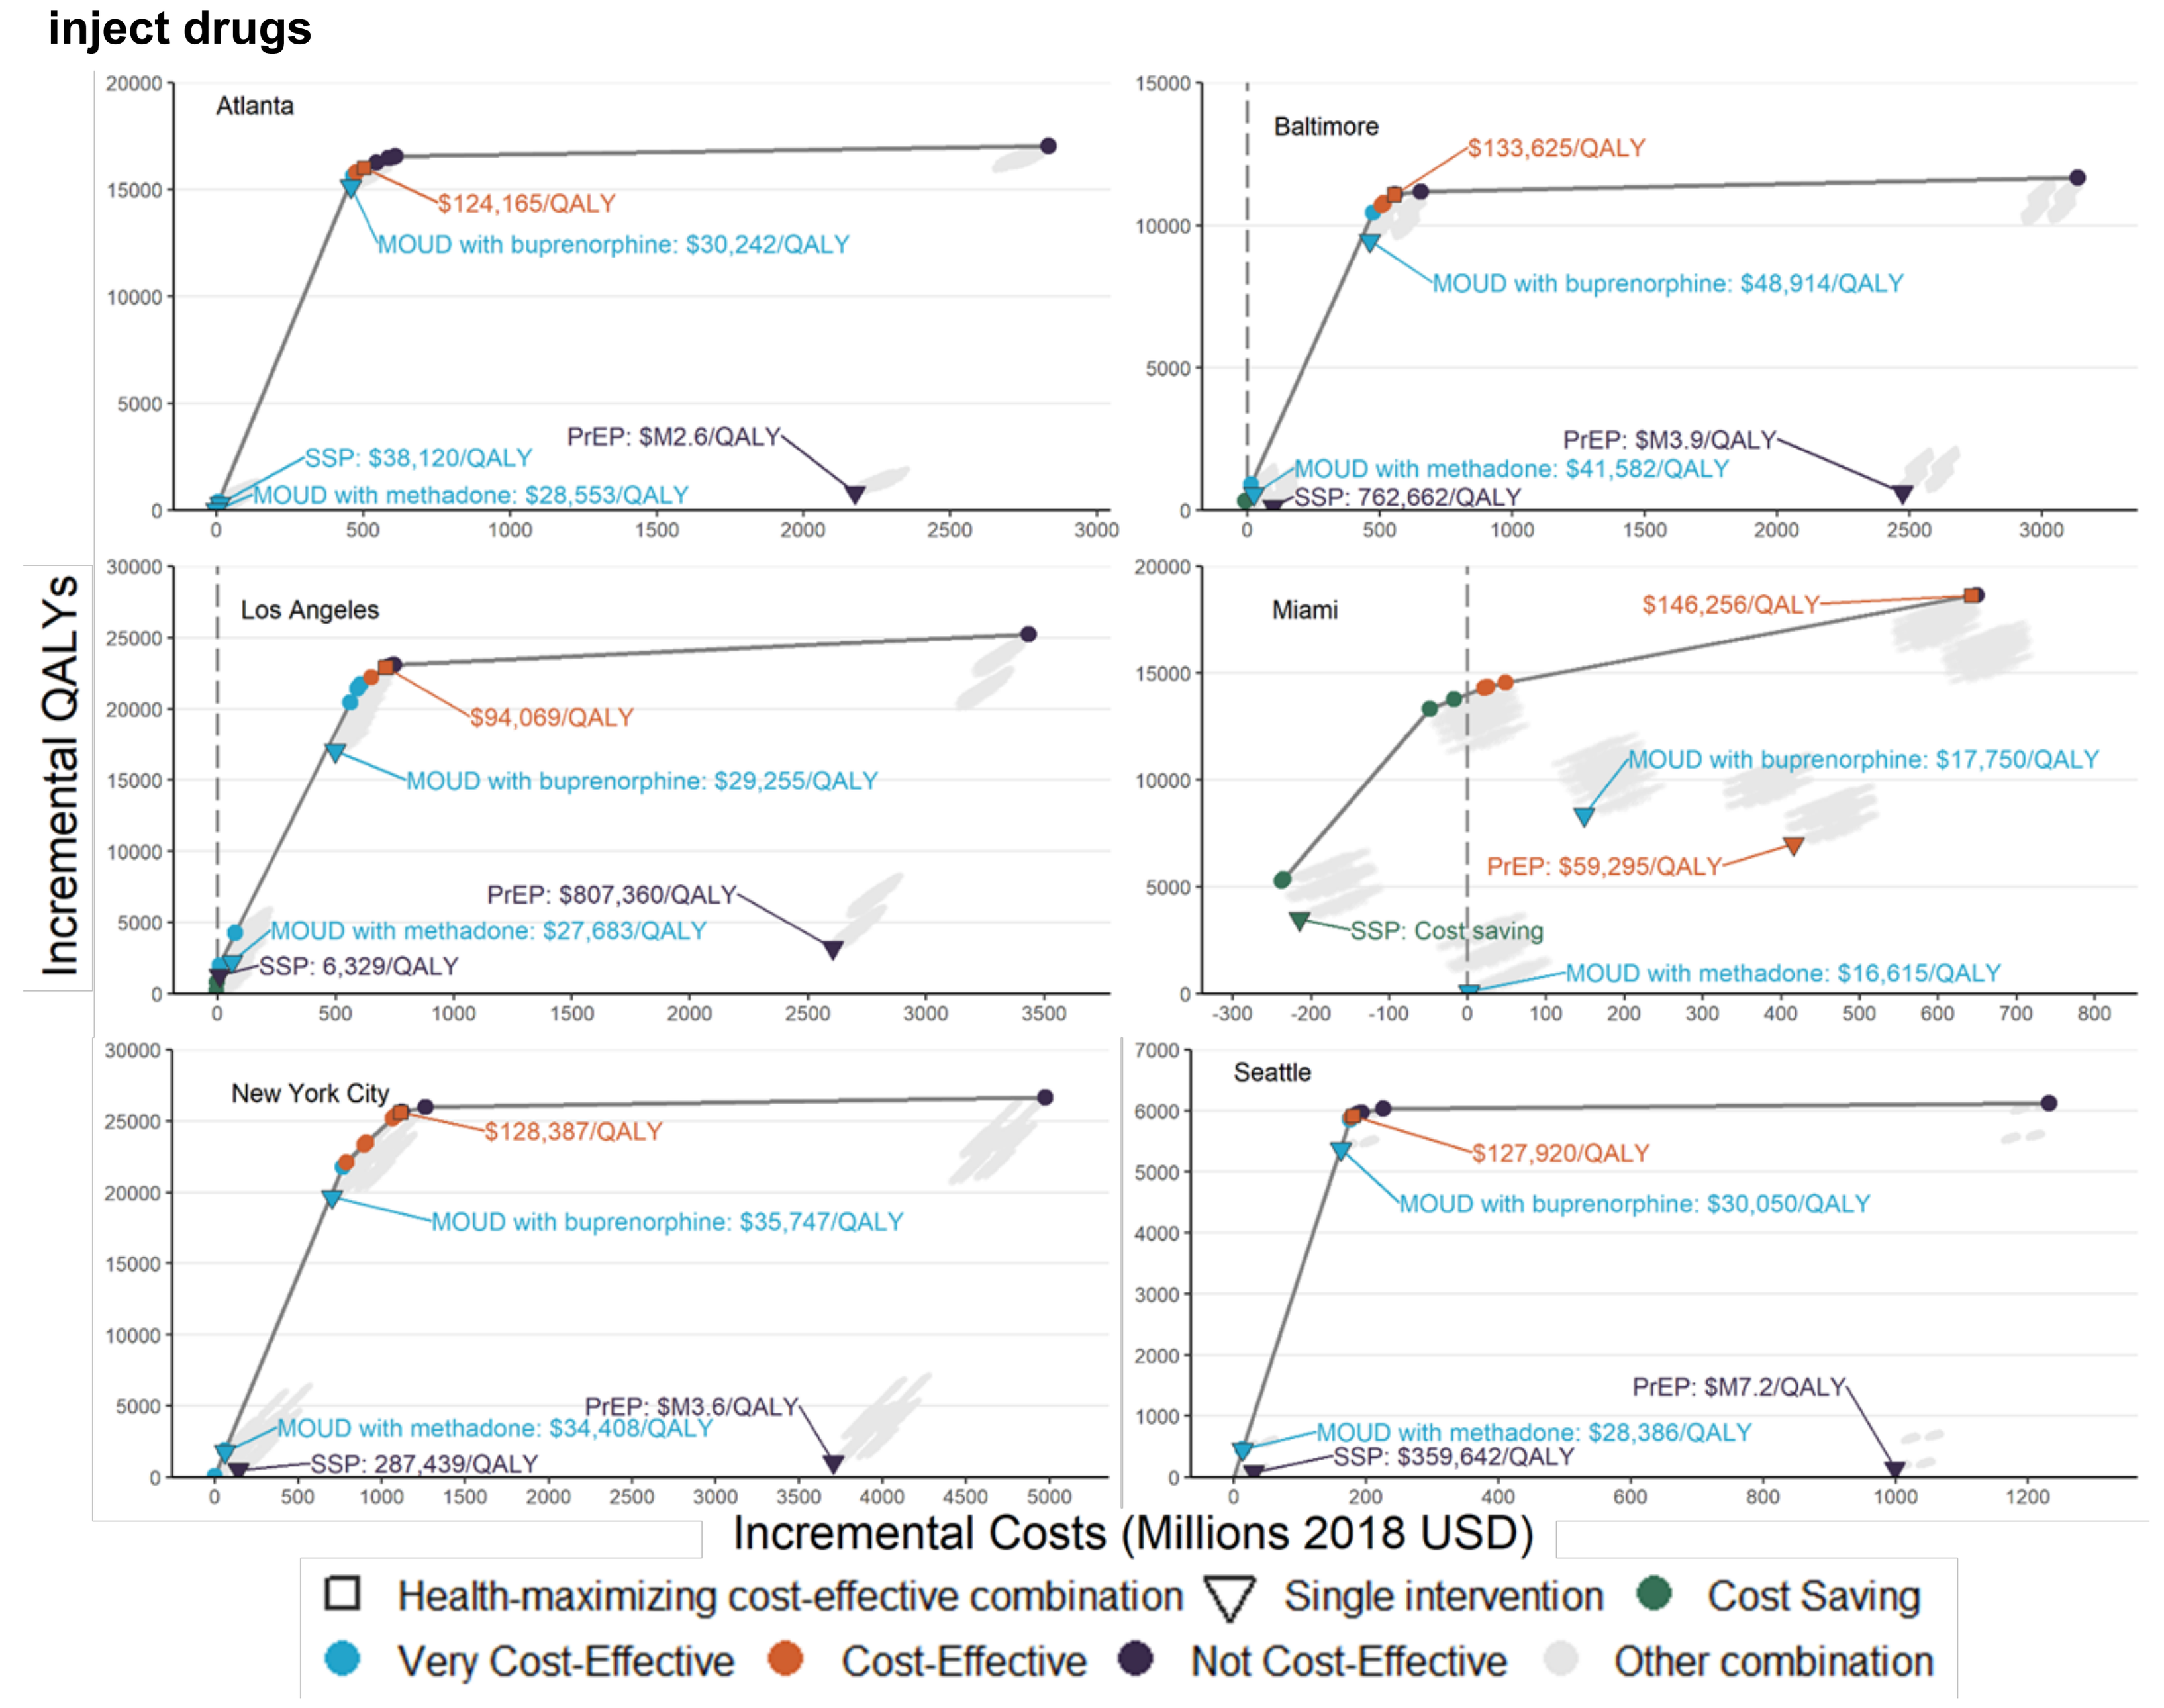
Incremental QALYs
Incremental Costs (Millions 2018 USD)
Atlanta
$124,165/QALY
MOUD with buprenorphine: $30,242/QALY
SSP: $38,120/QALY
MOUD with methadone: $28,553/QALY
PrEP: $M2.6/QALY
Baltimore
$133,625/QALY
MOUD with buprenorphine: $48,914/QALY
PrEP: $M3.9/QALY
MOUD with methadone: $41,582/QALY
SSP: 762,662/QALY
Los Angeles
$94,069/QALY
MOUD with buprenorphine: $29,255/QALY
PrEP: $807,360/QALY
MOUD with methadone: $27,683/QALY
SSP: 6,329/QALY
Miami
$146,256/QALY
MOUD with buprenorphine: $17,750/QALY
PrEP: $59,295/QALY
SSP: Cost saving
MOUD with methadone: $16,615/QALY
New York City
$128,387/QALY
MOUD with buprenorphine: $35,747/QALY
PrEP: $M3.6/QALY
MOUD with methadone: $34,408/QALY
SSP: 287,439/QALY
Seattle
$127,920/QALY
MOUD with buprenorphine: $30,050/QALY
PrEP: $M7.2/QALY
MOUD with methadone: $28,386/QALY
SSP: $359,642/QALY
Health-maximizing cost-effective combination
Single intervention
Cost Saving
Very Cost-Effective
Cost-Effective
Not Cost-Effective
Other combination

Figure 2. Projected reductions in HIV incidence among persons who inject drugs and men who have sex with men who inject drugs

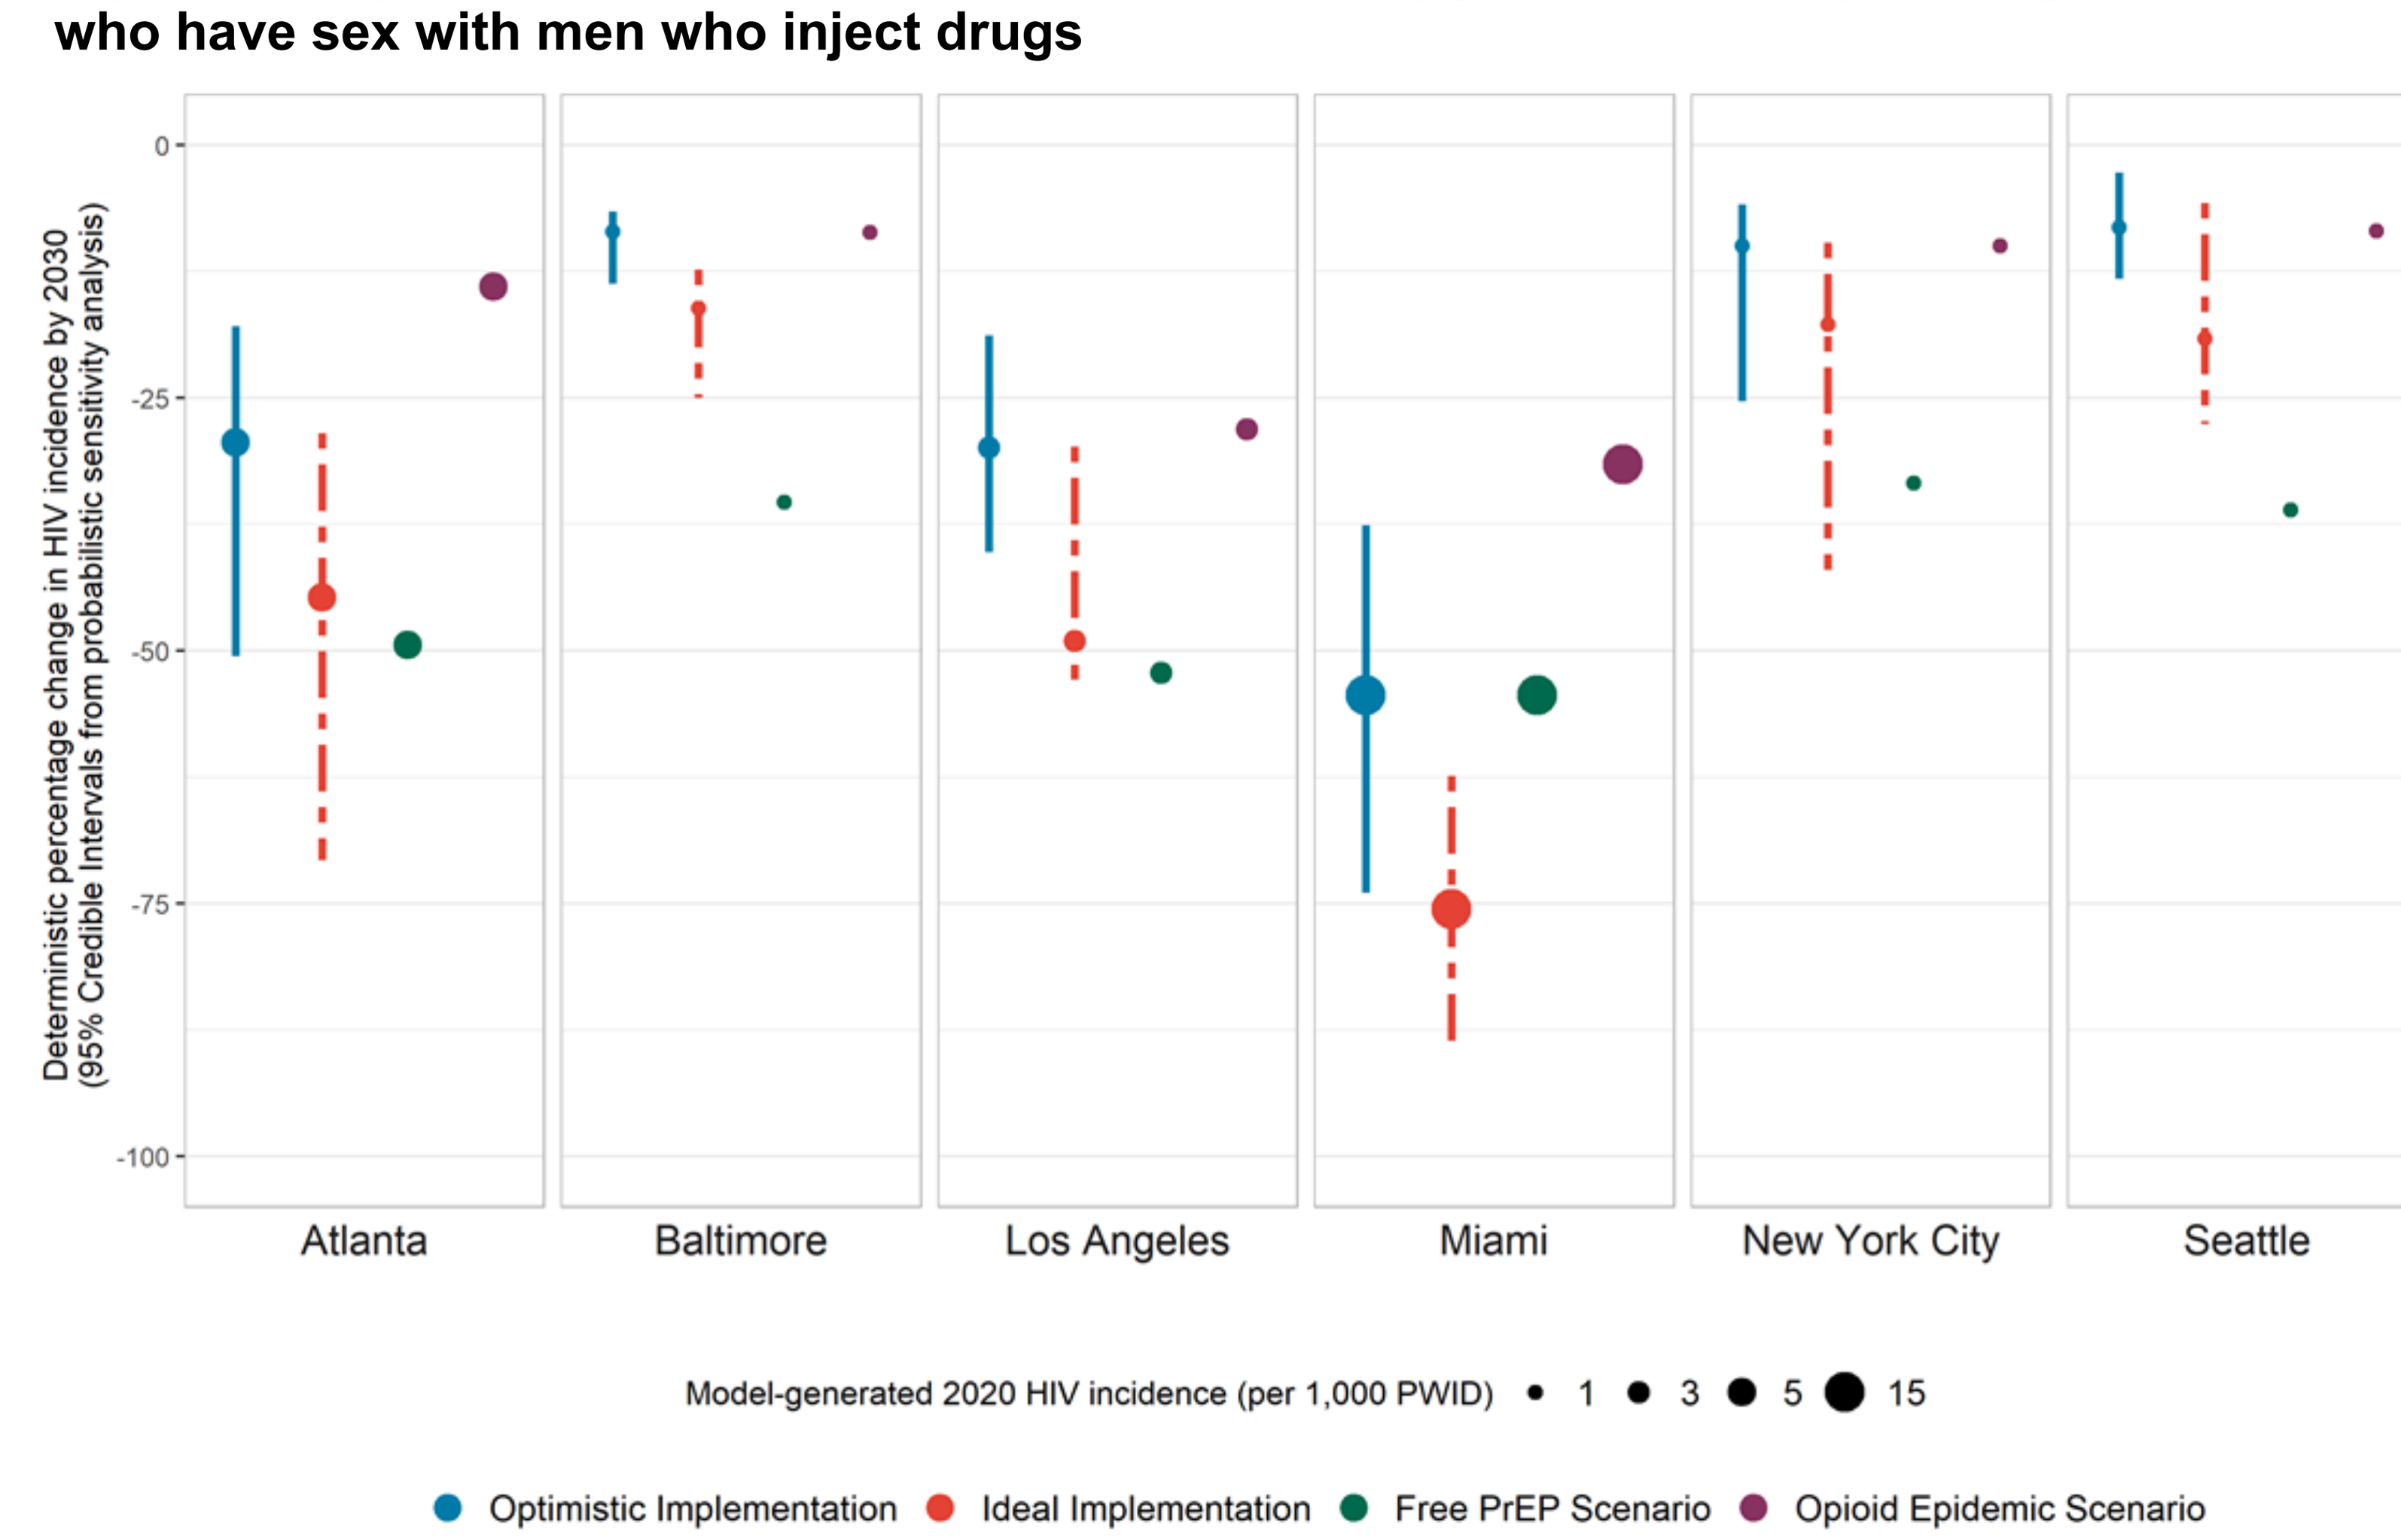
Deterministic percentage change in HIV incidence by 2030
(95% Credible Intervals from probabilistic sensitivity analysis)
Atlanta
Baltimore
Los Angeles
Miami
New York City
Seattle
Model-generated 2020 HIV incidence (per 1,000 PWID) 1 3 5 15
Optimistic Implementation
Ideal Implementation
Free PrEP Scenario
Opioid Epidemic Scenario