Rapid uptake of HIV pre-exposure prophylaxis (PrEP) in a publicly funded population-based program in British Columbia, Canada

K. Junine Toy[1], Jason Trigg[1], Wendy W. Zhang[1], Paul Sereda[1], Viviane Dias Lima[1], Katherine J. Lepik[1], Mark W. Hull[1], Raquel M. Espinoza[1], Silvia Guillemi[1], David Hall[2], David M. Moore[1], Rolando Barrios[1], Julio S.G. Montaner[1]

1. BC Centre for Excellence in HIV/AIDS, Vancouver, BC, Canada 2. Vancouver Coastal Health, Vancouver, BC, Canada

Abstract 956

Background

In January 2018, a 100% publicly funded population-based HIV PrEP program was launched in British Columbia (BC), Canada. Persons meeting BC PrEP program eligibility criteria qualified for daily emtricitabine-tenofovir DF PrEP at no cost through the centralized BC Centre for Excellence in HIV/AIDS (BC-CfE) Drug Treatment Program (Fig. 1). Here we describe program enrolment, client and prescriber characteristics, and preliminary participant outcomes including reported adverse drug reactions and PrEP discontinuation in the first year of the program.

Figure 1: BC PrEP Program Eligibility Criteria and the PrEP Enrolment & Prescription Request Form

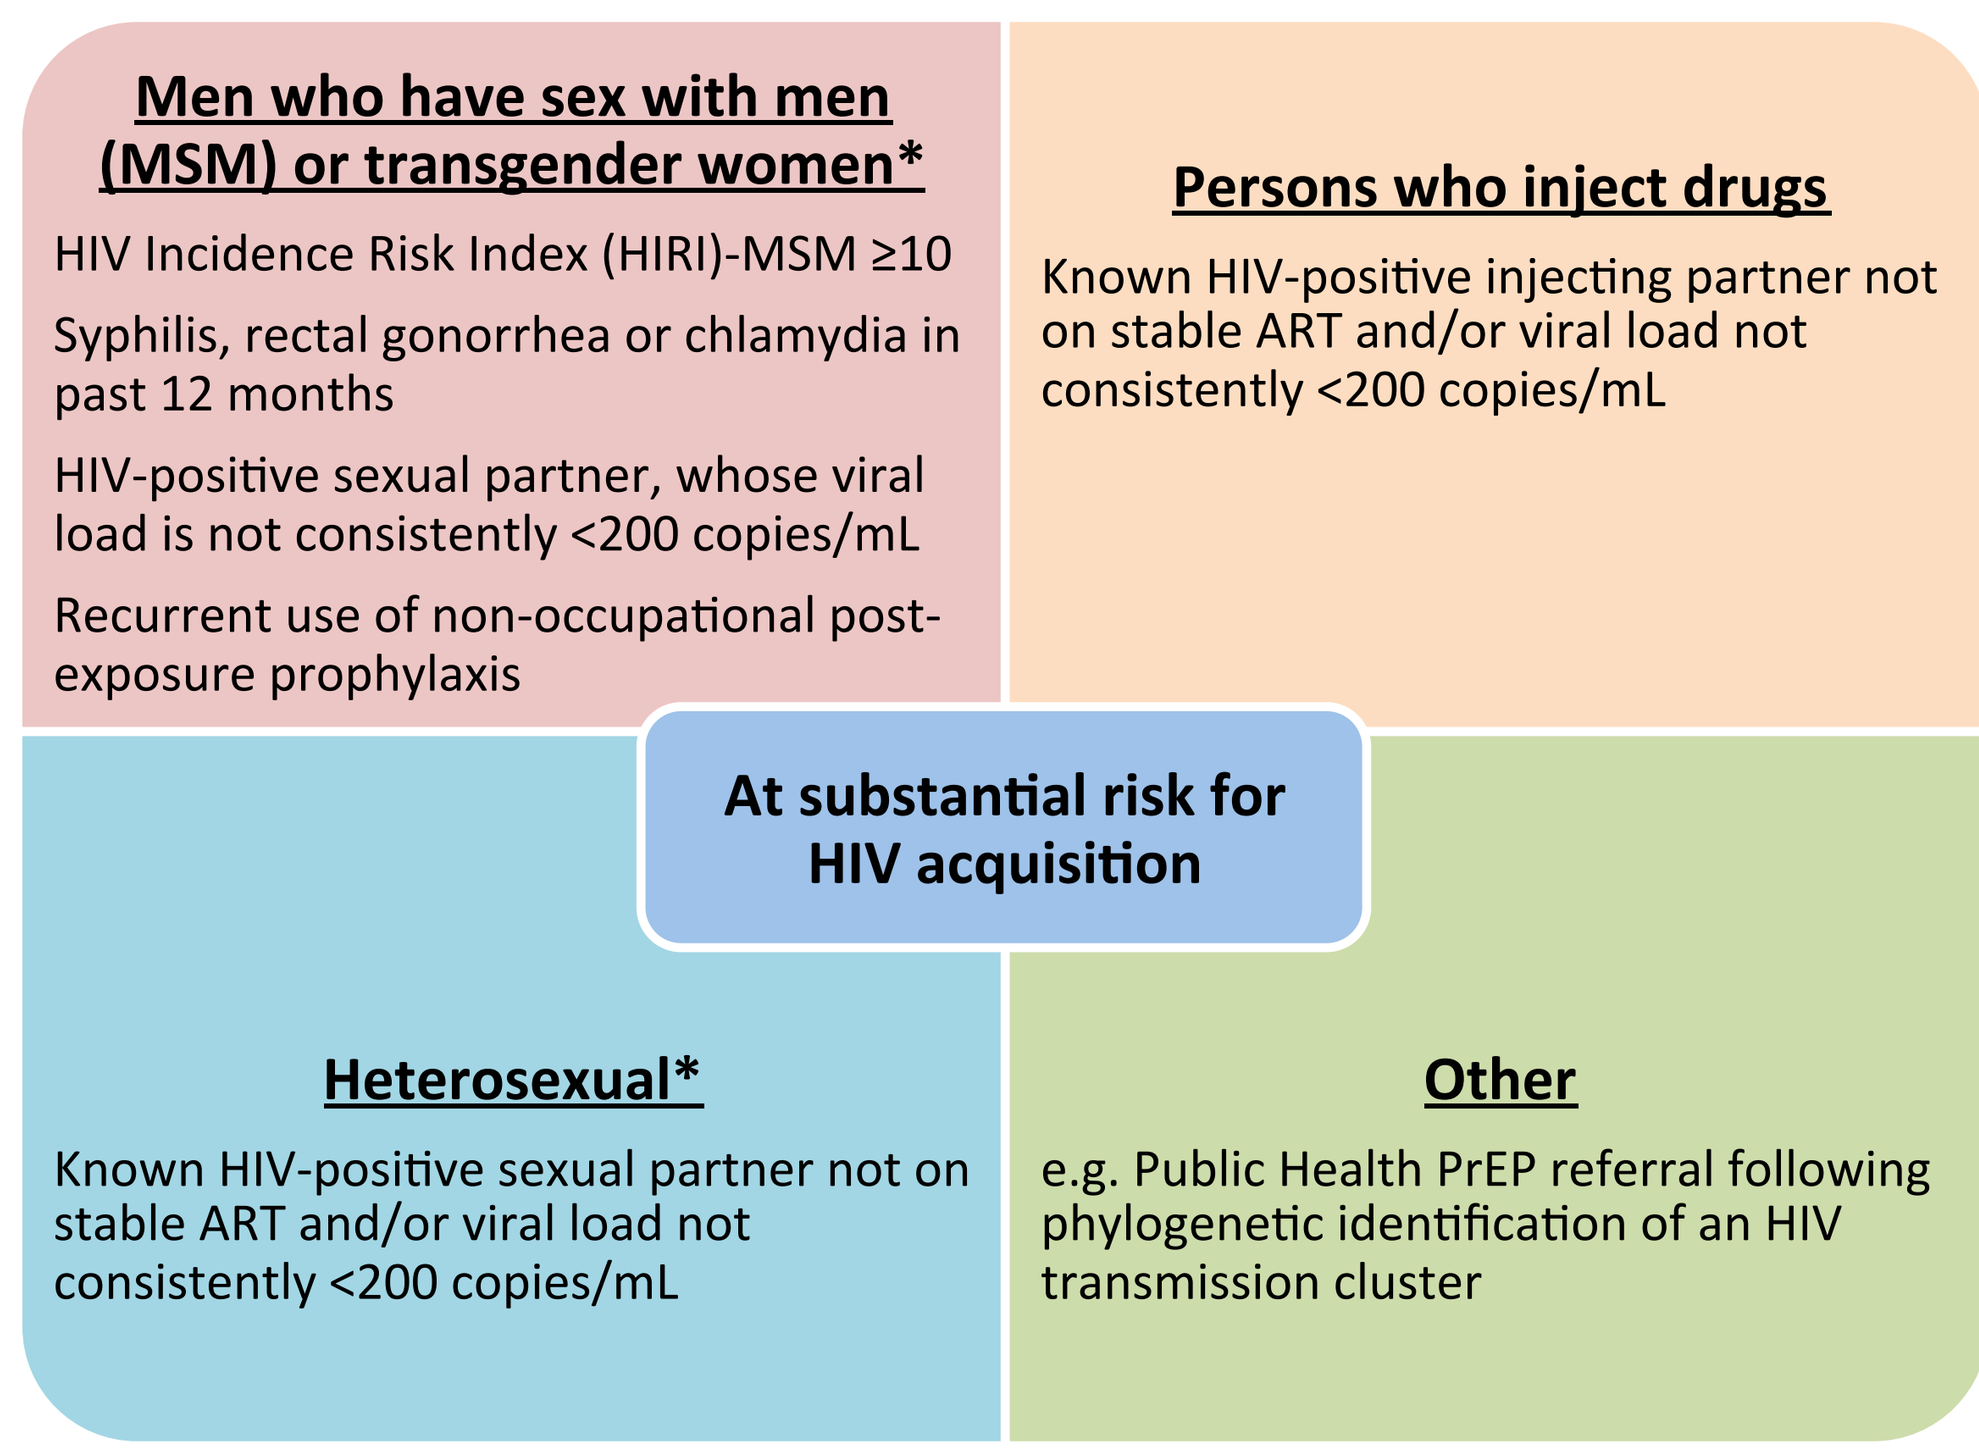
Men who have sex with men (MSM) or transgender women*
HIV Incidence Risk Index (HIRI)-MSM ≥10
Syphilis, rectal gonorrhea or chlamydia in past 12 months
HIV-positive sexual partner, whose viral load is not consistently <200 copies/mL
Recurrent use of non-occupational post-exposure prophylaxis
Persons who inject drugs
Known HIV-positive injecting partner not on stable ART and/or viral load not consistently <200 copies/mL
At substantial risk for HIV acquisition
Heterosexual*
Known HIV-positive sexual partner not on stable ART and/or viral load not consistently <200 copies/mL
Other
e.g. Public Health PrEP referral following phylogenetic identification of an HIV transmission cluster

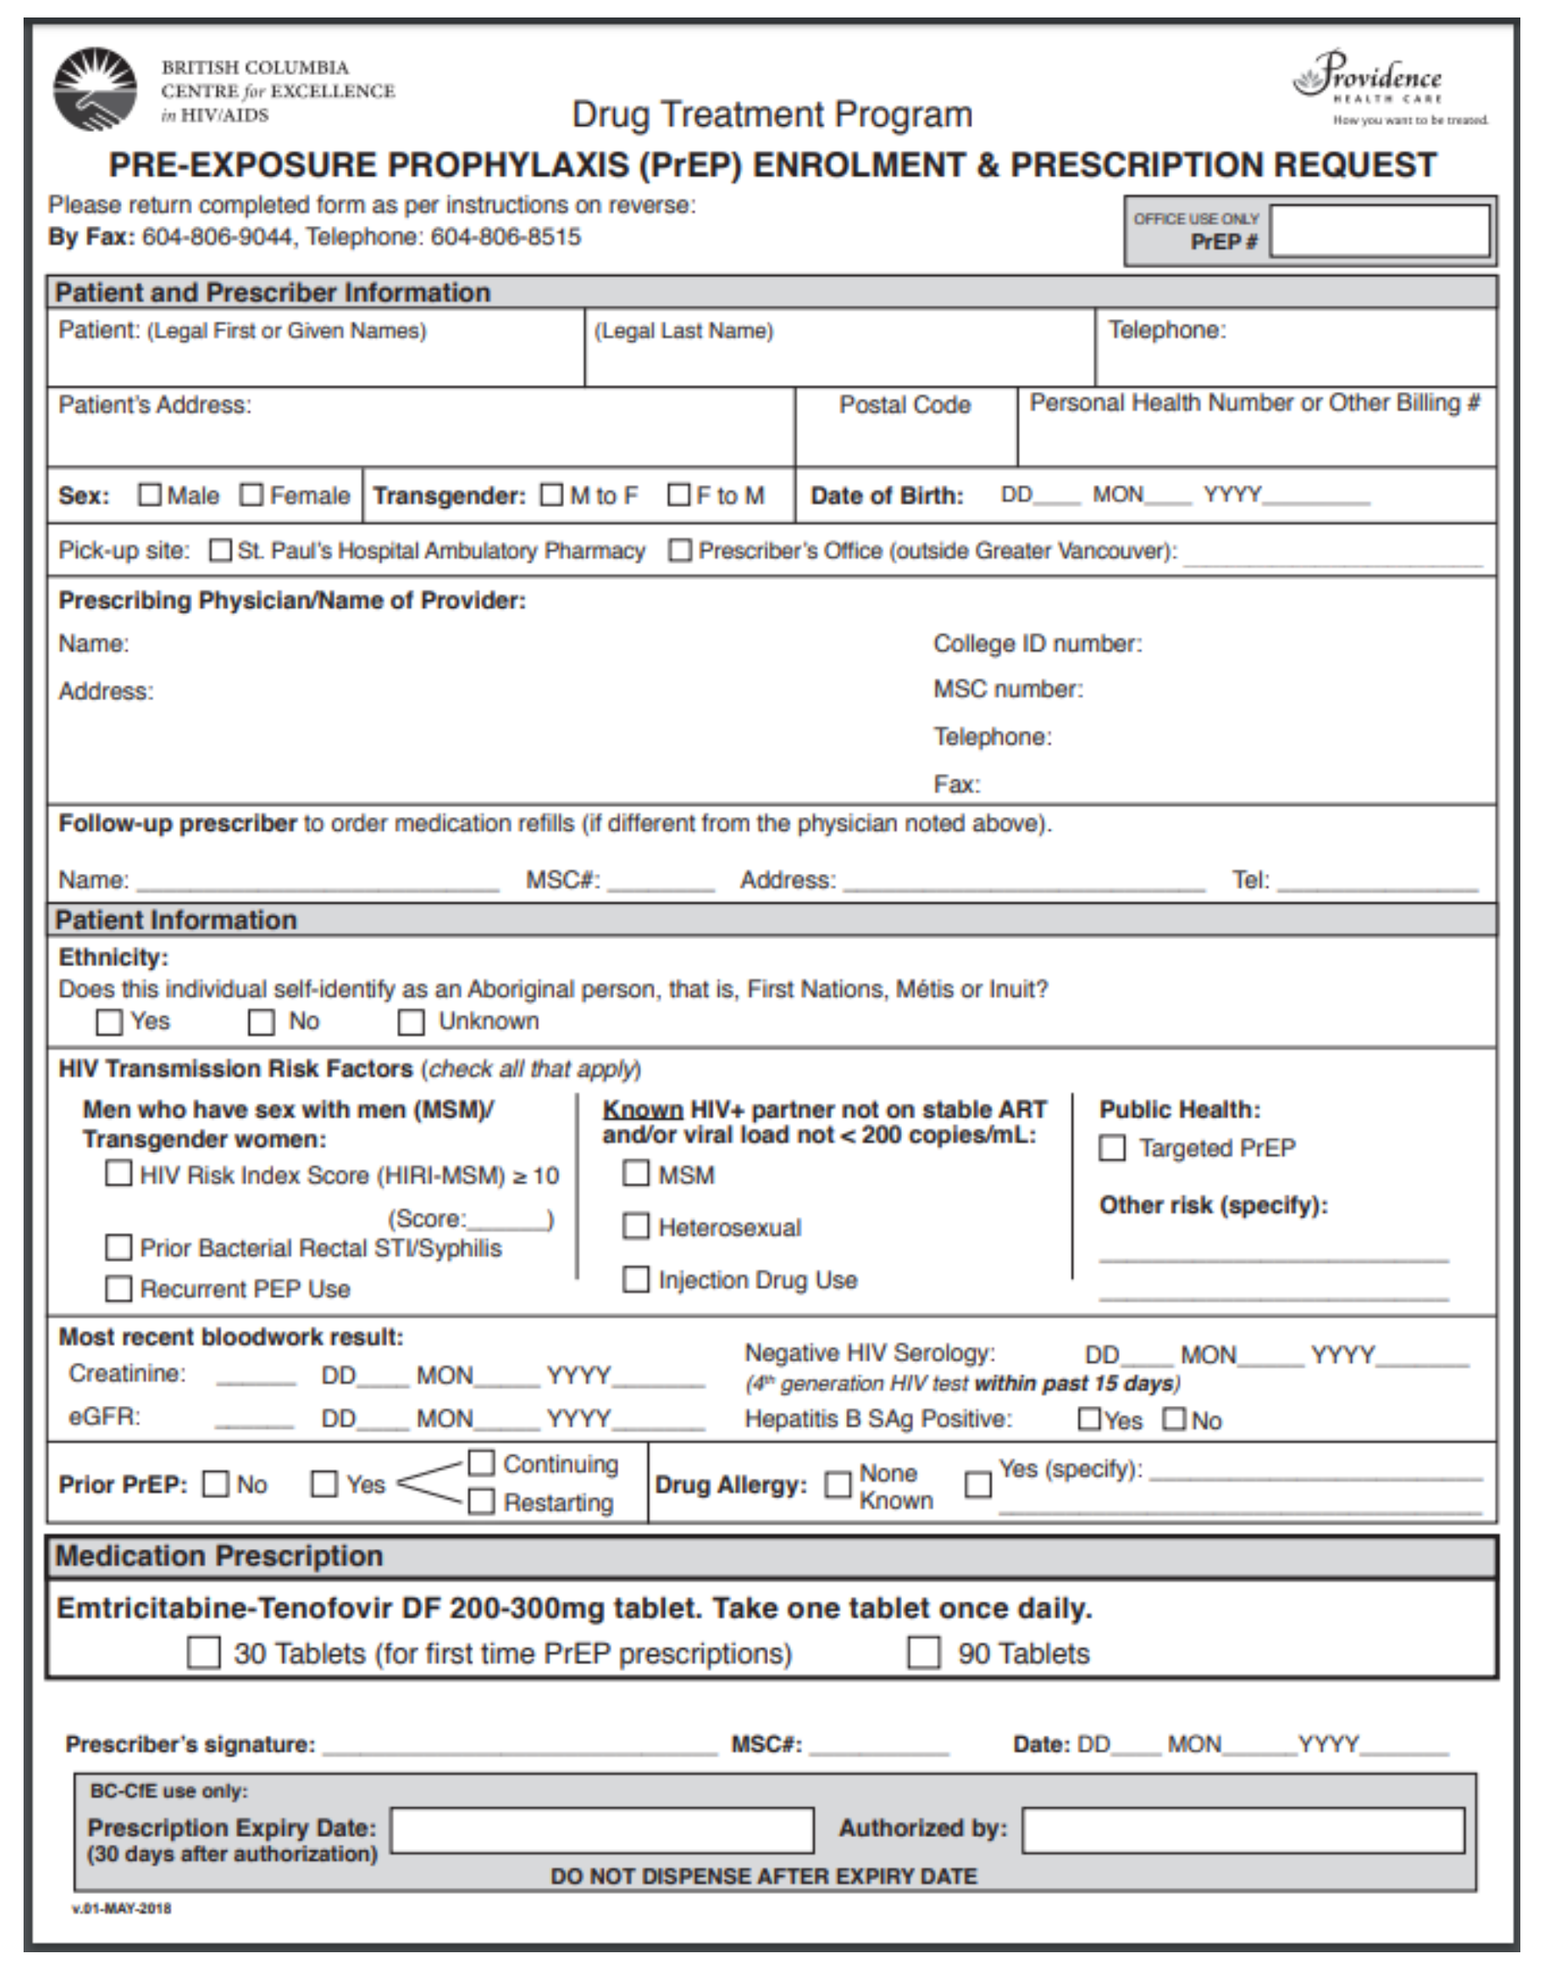
BRITISH COLUMBIA CENTRE for EXCELLENCE in HIV/AIDS — Providence Health Care

Drug Treatment Program

PRE-EXPOSURE PROPHYLAXIS (PrEP) ENROLMENT & PRESCRIPTION REQUEST

Please return completed form as per instructions on reverse:
By Fax: 604-806-9044, Telephone: 604-806-8515

OFFICE USE ONLY PrEP #

Patient and Prescriber Information
Patient: (Legal First or Given Names) | (Legal Last Name) | Telephone:
Patient's Address: | Postal Code | Personal Health Number or Other Billing #
Sex: ☐ Male ☐ Female Transgender: ☐ M to F ☐ F to M Date of Birth: DD___ MON___ YYYY________
Pick-up site: ☐ St. Paul's Hospital Ambulatory Pharmacy ☐ Prescriber's Office (outside Greater Vancouver):

Prescribing Physician/Name of Provider:
Name: College ID number:
Address: MSC number:
Telephone:
Fax:

Follow-up prescriber to order medication refills (if different from the physician noted above).
Name: ________ MSC#: ____ Address: ________ Tel: ______

Patient Information
Ethnicity:
Does this individual self-identify as an Aboriginal person, that is, First Nations, Métis or Inuit?
☐ Yes ☐ No ☐ Unknown

HIV Transmission Risk Factors (check all that apply)
Men who have sex with men (MSM)/ Transgender women: ☐ HIV Risk Index Score (HIRI-MSM) ≥ 10 (Score:_____) ☐ Prior Bacterial Rectal STI/Syphilis ☐ Recurrent PEP Use
Known HIV+ partner not on stable ART and/or viral load not < 200 copies/mL: ☐ MSM ☐ Heterosexual ☐ Injection Drug Use
Public Health: ☐ Targeted PrEP
Other risk (specify):

Most recent bloodwork result:
Creatinine: _____ DD___ MON____ YYYY_____
eGFR: _____ DD___ MON____ YYYY_____
Negative HIV Serology: DD___ MON____ YYYY_____ (4th generation HIV test within past 15 days)
Hepatitis B SAg Positive: ☐ Yes ☐ No

Prior PrEP: ☐ No ☐ Yes ☐ Continuing ☐ Restarting Drug Allergy: ☐ None Known ☐ Yes (specify): ______

Medication Prescription
Emtricitabine-Tenofovir DF 200-300mg tablet. Take one tablet once daily.
☐ 30 Tablets (for first time PrEP prescriptions) ☐ 90 Tablets

Prescriber's signature: ________ MSC#: ______ Date: DD___ MON____ YYYY_____
BC-CfE use only:
Prescription Expiry Date: (30 days after authorization) Authorized by:
DO NOT DISPENSE AFTER EXPIRY DATE

v.01-MAY-2018

* Reporting condomless intercourse

Must also meet all of the following criteria for PrEP program eligibility:
- BC resident and provincial healthcare coverage
- Creatinine clearance or estimated glomerular filtration rate >60 mL/min/1.73 m^2
- Documented hepatitis B infection status
- Current negative 4th generation HIV antigen/antibody test (within 15 days of application)

Methods

- Clients for whom PrEP was requested through the BC program from 1-Jan-2018 to 31-Dec-2018 (with follow-up until 15-Feb-2019) were described by clinical and demographic characteristics.
- Prescribers were summarized by practice setting and HIV management experience.
- Comparisons between prescriber settings for PrEP enrolment and days from baseline HIV test to PrEP dispensing used Chi-Squared test for categorical and Wilcoxon rank sum test for continuous variables.
- Reported reasons for PrEP discontinuation and adverse drug reactions (ADR) were summarized.

Links:
- BC Centre for Excellence in HIV/AIDS. PrEP Program Information. Found at http://cfenet.ubc.ca/hiv-pre-exposure-prophylaxis-prep
- BC Centre for Excellence in HIV/AIDS. Guidance for the use of Pre-exposure prophylaxis (PrEP) for the prevention of HIV acquisition in British Columbia. October 2017. Found at http://www.cfenet.ubc.ca/publications/centre-documents/guidance-for-the-use-pre-exposure-prophylaxis-prep-prevention-hiv-acquisition

Acknowledgements: The authors would like to acknowledge Ms. Jelena Antic and the BC-CfE Drug Treatment Program staff, and Mr. Jack da Silva and St. Paul's Hospital Ambulatory Pharmacy for their valued support for the BC PrEP program.

CROI Conference on Retroviruses and Opportunistic Infections

Correspondence to: Dr. Julio Montaner jmontaner@cfenet.ubc.ca

Results

- ***Eligible clients:*** In the first year, there were 3351 PrEP applicants, 3284 (98%) of whom met eligibility criteria (Table 1). Most (84%) resided in Greater Vancouver (Fig. 2) and 2.3% in a rural location.
- Approved clients were 98% male and median age 34 years (range 17-81) (Table 2). Most (81%) were PrEP-naïve at enrolment; the remainder transferred from client-paid or private insurance coverage.
- Mean baseline estimated glomerular filtration rate (eGFR) was 94 mL/min/1.73 m^2 (SD 16.8) in PrEP experienced vs. 100 mL/min/1.73 m^2 (SD 16.8) in PrEP naïve participants (p<0.001).

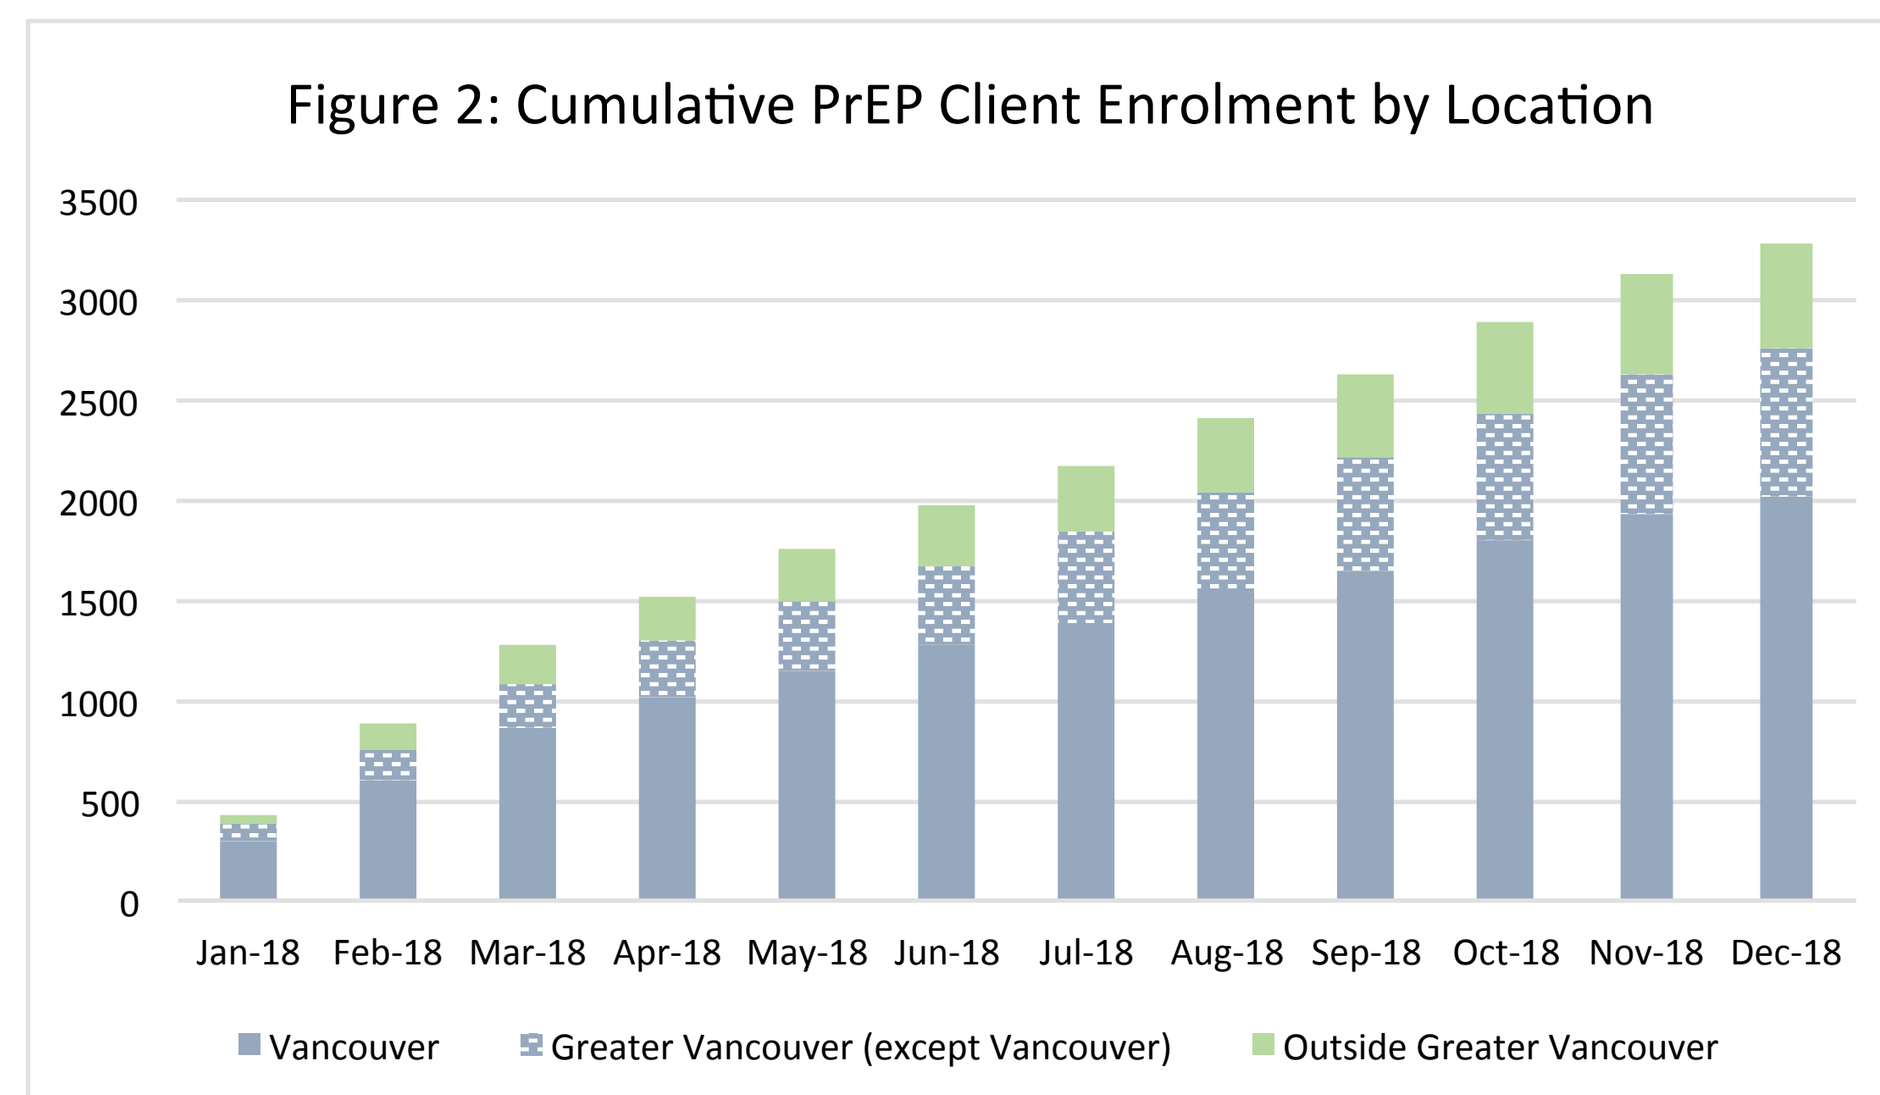
3500
3000
2500
2000
1500
1000
500
0
Jan-18
Feb-18
Mar-18
Apr-18
May-18
Jun-18
Jul-18
Aug-18
Sep-18
Oct-18
Nov-18
Dec-18
Vancouver
Greater Vancouver (except Vancouver)
Outside Greater Vancouver
Figure 2: Cumulative PrEP Client Enrolment by Location

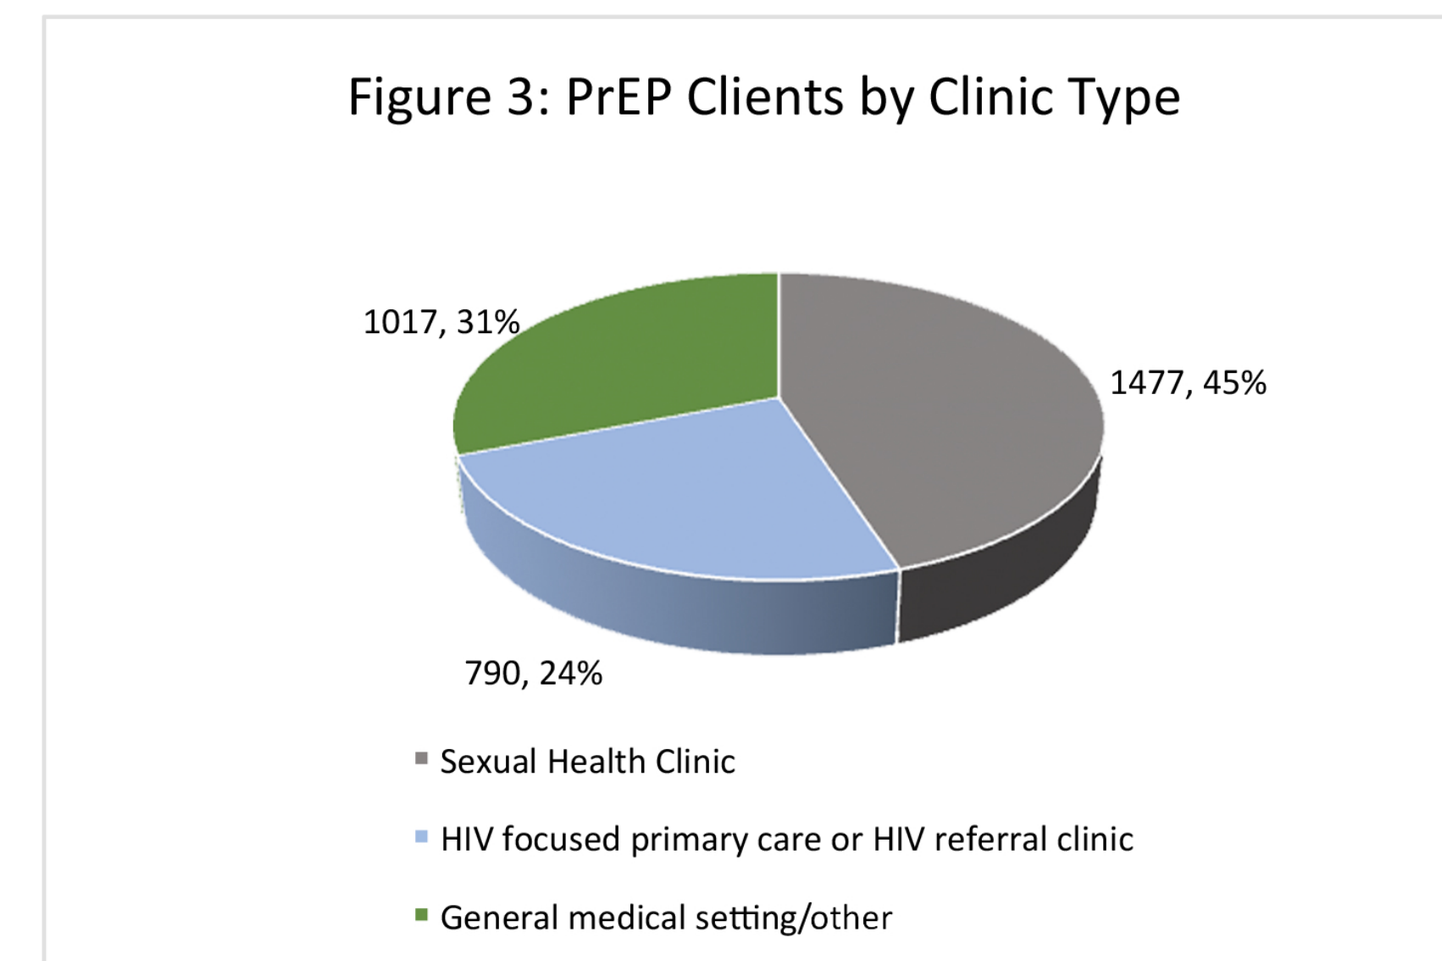
1017, 31%
1477, 45%
790, 24%
Sexual Health Clinic
HIV focused primary care or HIV referral clinic
General medical setting/other
Figure 3: PrEP Clients by Clinic Type

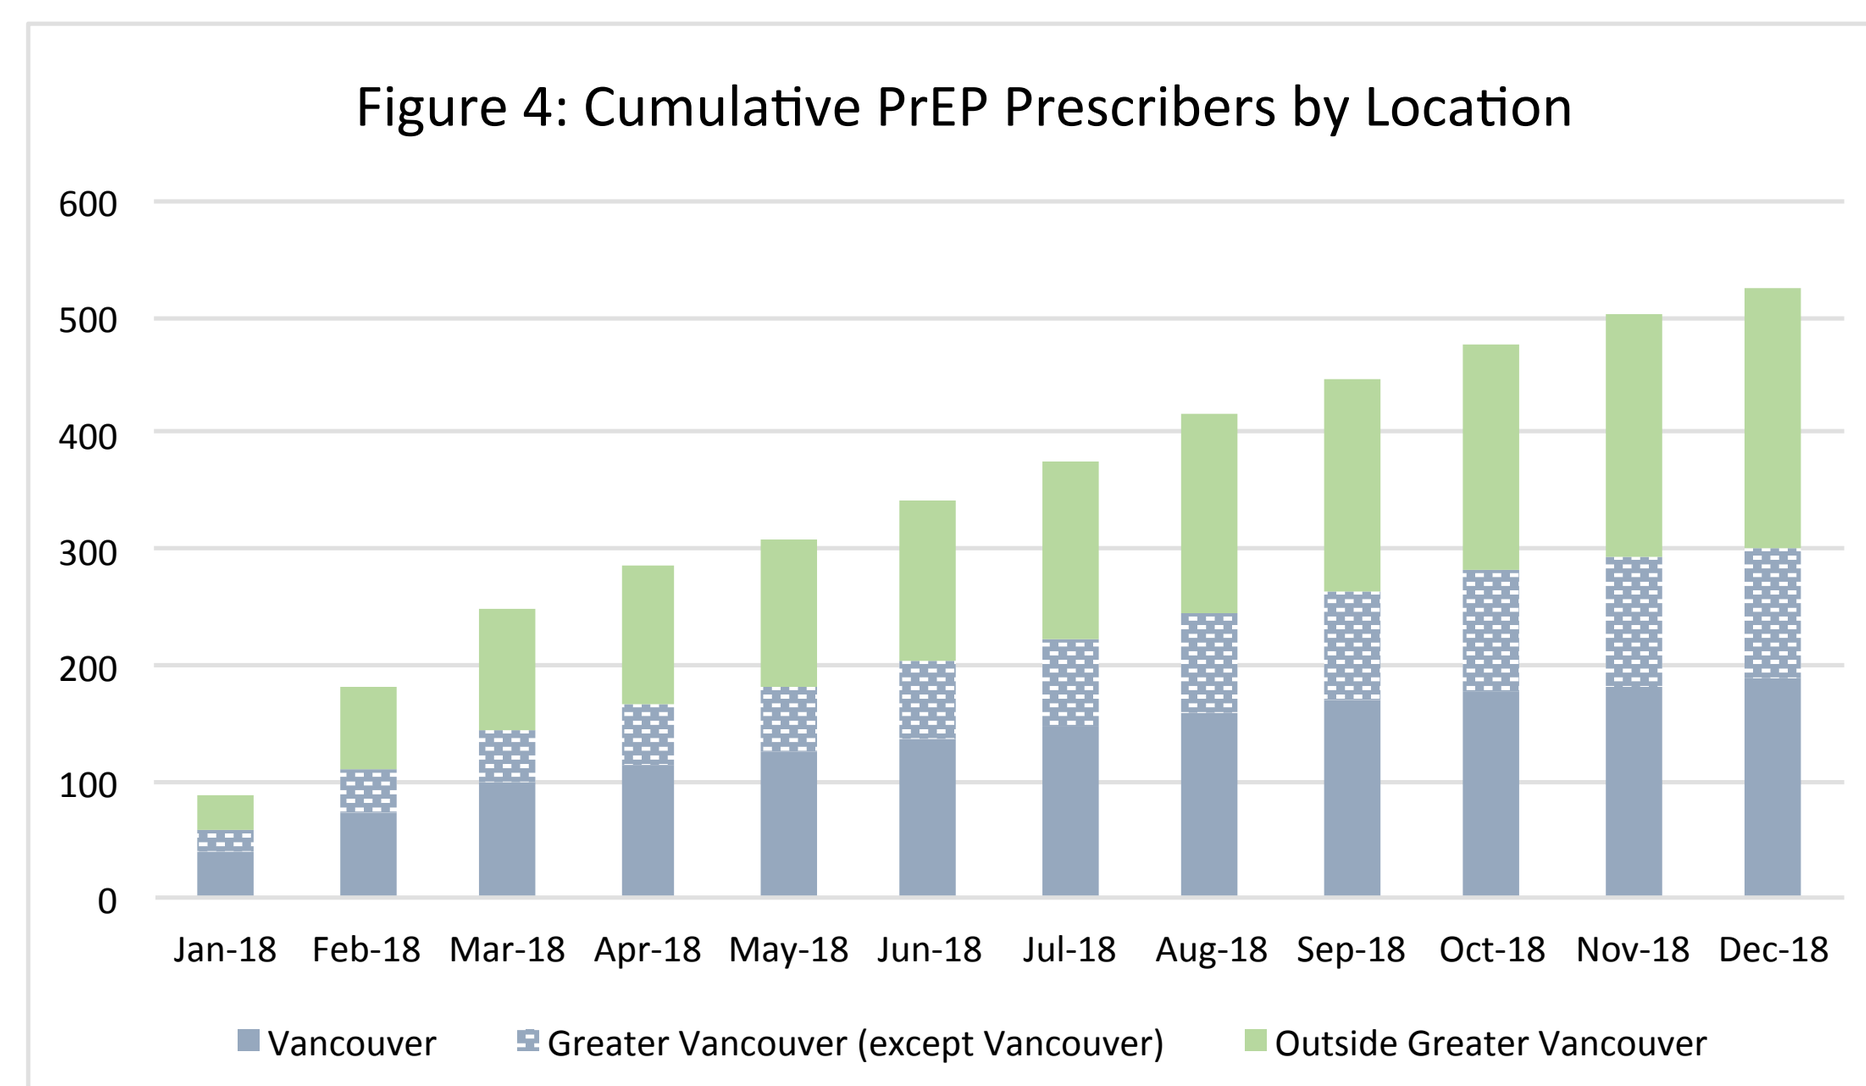
600
500
400
300
200
100
0
Jan-18
Feb-18
Mar-18
Apr-18
May-18
Jun-18
Jul-18
Aug-18
Sep-18
Oct-18
Nov-18
Dec-18
Vancouver
Greater Vancouver (except Vancouver)
Outside Greater Vancouver
Figure 4: Cumulative PrEP Prescribers by Location

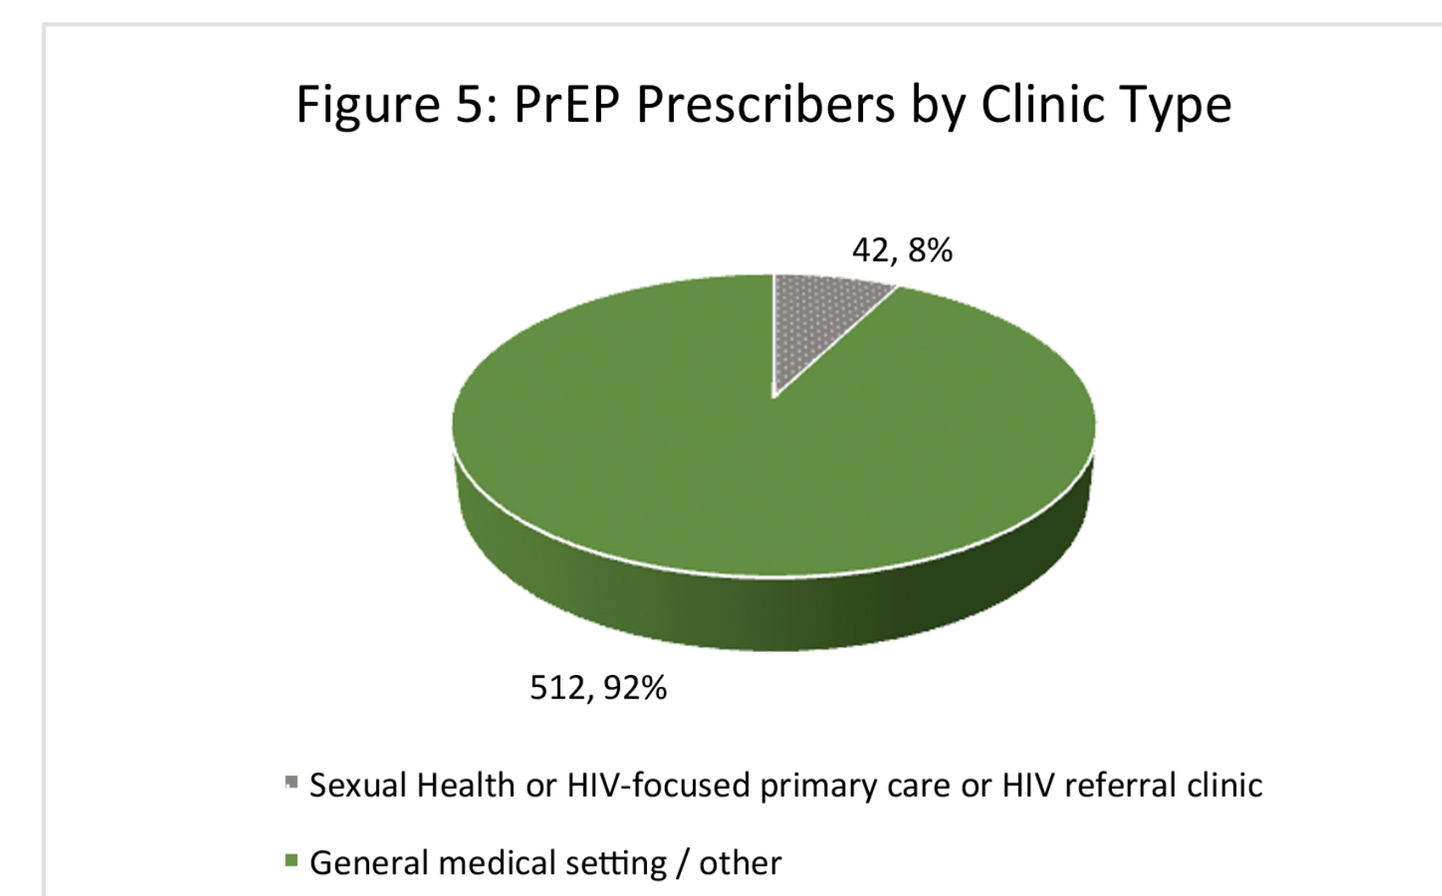
42, 8%
512, 92%
Sexual Health or HIV-focused primary care or HIV referral clinic
General medical setting / other
Figure 5: PrEP Prescribers by Clinic Type

- ***Prescribers:*** There were 554 enrolling PrEP prescribers (Fig. 4-5), of whom 289 (52%) had no previous HIV care and treatment experience in BC. Nurse practitioners (n=12) provided follow-up care to 8.6% of PrEP clients.
- Prescribers at sexual health, HIV-focused primary care or HIV referral clinics (n=42) had median (Q1-Q3) 21 (5-46) PrEP clients each vs. 1 (1-2) clients for the 512 prescribers in general or other practice settings (p<0.001).
- ***Prescriptions:*** Time from baseline HIV negative test to first PrEP dispensing was median 10 (7-13) days for clients of sexual health or HIV clinics vs. 10 (8-14) days at other practice settings (p<0.001).
- Although BC guidelines recommend daily PrEP, intermittent or on-demand use was documented for 103 clients (3.1%).
- PrEP discontinuation or lapsed prescription was noted in 291 clients (8.9%) (Table 3). Adverse drug reaction was reported in 28 PrEP clients (<1%) (Table 4), 19 of whom discontinued medication.
- Two cases of new HIV infection were diagnosed prior to end of follow-up (<0.1%); both occurred in individuals who had discontinued PrEP.

Conclusion

In the first year following launch of a publicly-funded PrEP program in BC, almost 3300 clients were enrolled by >550 prescribers. Early participants were predominantly at-risk MSM residing in urban areas. To date there have been few reports of PrEP discontinuation or seroconversion.

Table 1: Total PrEP Applications (1 –Jan-2018 to 31-Dec-2018)	N=3351
Persons not meeting program eligibility criteria	**N=67**
	n (%)
Not at high risk for HIV acquisition per program criteria	19 (28)
Not a resident of BC / no BC healthcare coverage	24 (36)
Qualifying blood work not provided	20 (30)
Compromised renal function: eGFR <60 mL/min/1.73 m^2	4 (6)
Persons meeting program eligibility criteria	**N=3284**
Program Qualifying Risk Factor(s) for HIV Acquisition:*	n (%)
Men who have sex with men, transgender women	3253 (99.1)
HIV Incidence Risk Index for MSM (HIRI-MSM) 10 - 24	2060 (62.7)
≥25	830 (25.3)
Infectious syphilis or rectal bacterial sexually transmitted infection (STI)	689 (21.0)
HIRI-MSM ≥10 and infectious syphilis or rectal bacterial STI	556 (16.9)
HIV-positive sexual partner**	206 (6.3)
Recurrent non-occupational post-exposure prophylaxis (NPEP) use	110 (3.4)
Heterosexual men and women who have an HIV-positive sexual partner**	19 (0.6)
Persons who inject drugs who have an HIV-positive injecting partner**	7 (0.2)
Public health referral following phylogenetic identification of an HIV cluster	27 (0.8)
Other risk factors	24 (0.7)

* more than one risk factor per client may be reported
** partner not receiving stable antiretroviral therapy and/or viral load not < 200 copies/mL

Table 2: Baseline characteristics of enrolled PrEP clients	N=3284
Median age (Q1-Q3), years	34 (28-45)
Gender identity, n (%)	
Male	3232 (98.4)
Female	16 (0.5)
Transgender female	22 (0.7)
Transgender male	10 (0.3)
Other gender identity, or unspecified	4 (0.1)
PrEP naïve at enrolment, n (%)	2651 (81.0)
Hep B surface antigen positive, n (%)	30 (0.9)
Mean eGFR (SD), mL/min/1.73 m^2	99 (16.9)

Table 3: PrEP patient and prescription status at end of follow-up period *	N=3284
	n (%)
PrEP discontinuation reported	88 (2.7)
No longer at risk / client choice	61 (1.8)
Adverse drug reaction	19 (0.6)
Other	2 (0.1)
Unspecified	11 (0.3)
>6 months since last PrEP prescription	203 (6.2)
HIV-positive seroconversion**	2 (0.06)
Deceased	1 (0.03)
Moved out of province	10 (0.3)

* Follow-up until 15-Feb-2019
** Persons not on PrEP at time of HIV diagnosis

Table 4: Adverse drug reactions reported (N=28 clients)

Symptom Category	n*	Detail
Gastrointestinal	13	diarrhea (6), nausea and/or vomiting (5), general GI upset or pain (6)
CNS	5	dizziness (2), headache (3), insomnia (1)
Dermatologic	5	rash (2), eczema (1), acne (1), flushing (1)
Renal	5	renal impairment (4), proteinuria (1)
General symptoms	4	fatigue/ malaise (4)
Other/ unspecified	6	neutropenia (1), peripheral neuropathy (1), dysuria (1), unspecified (3)

* Each adverse reaction report may include multiple symptom categories, and more than one symptom type within each symptom category
3 persons attributed their symptoms to a switch to a generic version of the emtricitabine-tenofovir tablet

BRITISH COLUMBIA CENTRE *for* EXCELLENCE *in* HIV/AIDS

Providence HEALTH CARE How you want to be treated.